Efficacy and safety of entinostat (ENT) and pembrolizumab (PEMBRO) in patients with non-small cell lung cancer (NSCLC) previously treated with anti-PD-(L)1 therapy.

Leena Gandhi[1], Pasi A. Jänne[2], Mateusz Opyrchal[3], Suresh S. Ramalingam[4], Igor I. Rybkin[5], Navid Hafez[6], Luis E. Raez[7], Dmitry Gabrilovich[8], Fang Wang[8], Peter Ordentlich[9], Susan Brouwer[9], Serap Sankoh[9], Emmett, V. Schmidt[10], Michael L. Meyers[11], Matthew D. Hellmann[12]

[1]NYU Perlmutter Cancer Center, New York, NY; [2]Dana-Farber Cancer Institute, Boston, MA; [3]Roswell Park Comprehensive Cancer Center, Buffalo, NY; [4]Winship Cancer Institute, Emory University, Atlanta, GA; [5]Josephine Ford Cancer Institute, Henry Ford Health System, Detroit, MI; [6]Yale Cancer Center, New Haven, CT; [7]Thoracic Oncology Program, Memorial Cancer Institute, Memorial Health Care System, Florida International University, Miami, FL; [8]The Wistar Institute, Philadelphia, PA; [9]Syndax Pharmaceuticals, Inc., Waltham, MA; [10]Merck & Co., Inc., Kenilworth, NJ; [11]Syndax Pharmaceuticals, Inc., New York, NY; [12]Memorial Sloan Kettering Cancer Center, New York, NY. For inquiries regarding this poster, please email: hellmanm@mskcc.org

BACKGROUND

Treatment options are limited in patients who progress on anti-PD-(L)1 therapy[1]. HDAC inhibitors have the potential to modulate myeloid-derived suppressor cell (MDSC) function and may synergize with PD-(L)1 inhibition[2].

Entinostat (ENT), an oral, class I-selective histone deacetylase inhibitor, enhances anti-PD-1 activity by downregulation of immunosuppressive cell types in the tumor microenvironment in vivo and has shown promising activity with pembrolizumab (PEMBRO) in patients with melanoma and lung cancer[3-5]. **Figure 1** describes the targets of entinostat and pembrolizumab in modulating immunosuppression.

We report here the preliminary results of a Phase 2 trial of entinostat plus pembrolizumab in patients with NSCLC previously treated with anti-PD-(L)1 therapy.

Figure 1. **Entinostat and Pembrolizumab Synergize to Modulate Immunosuppression.**

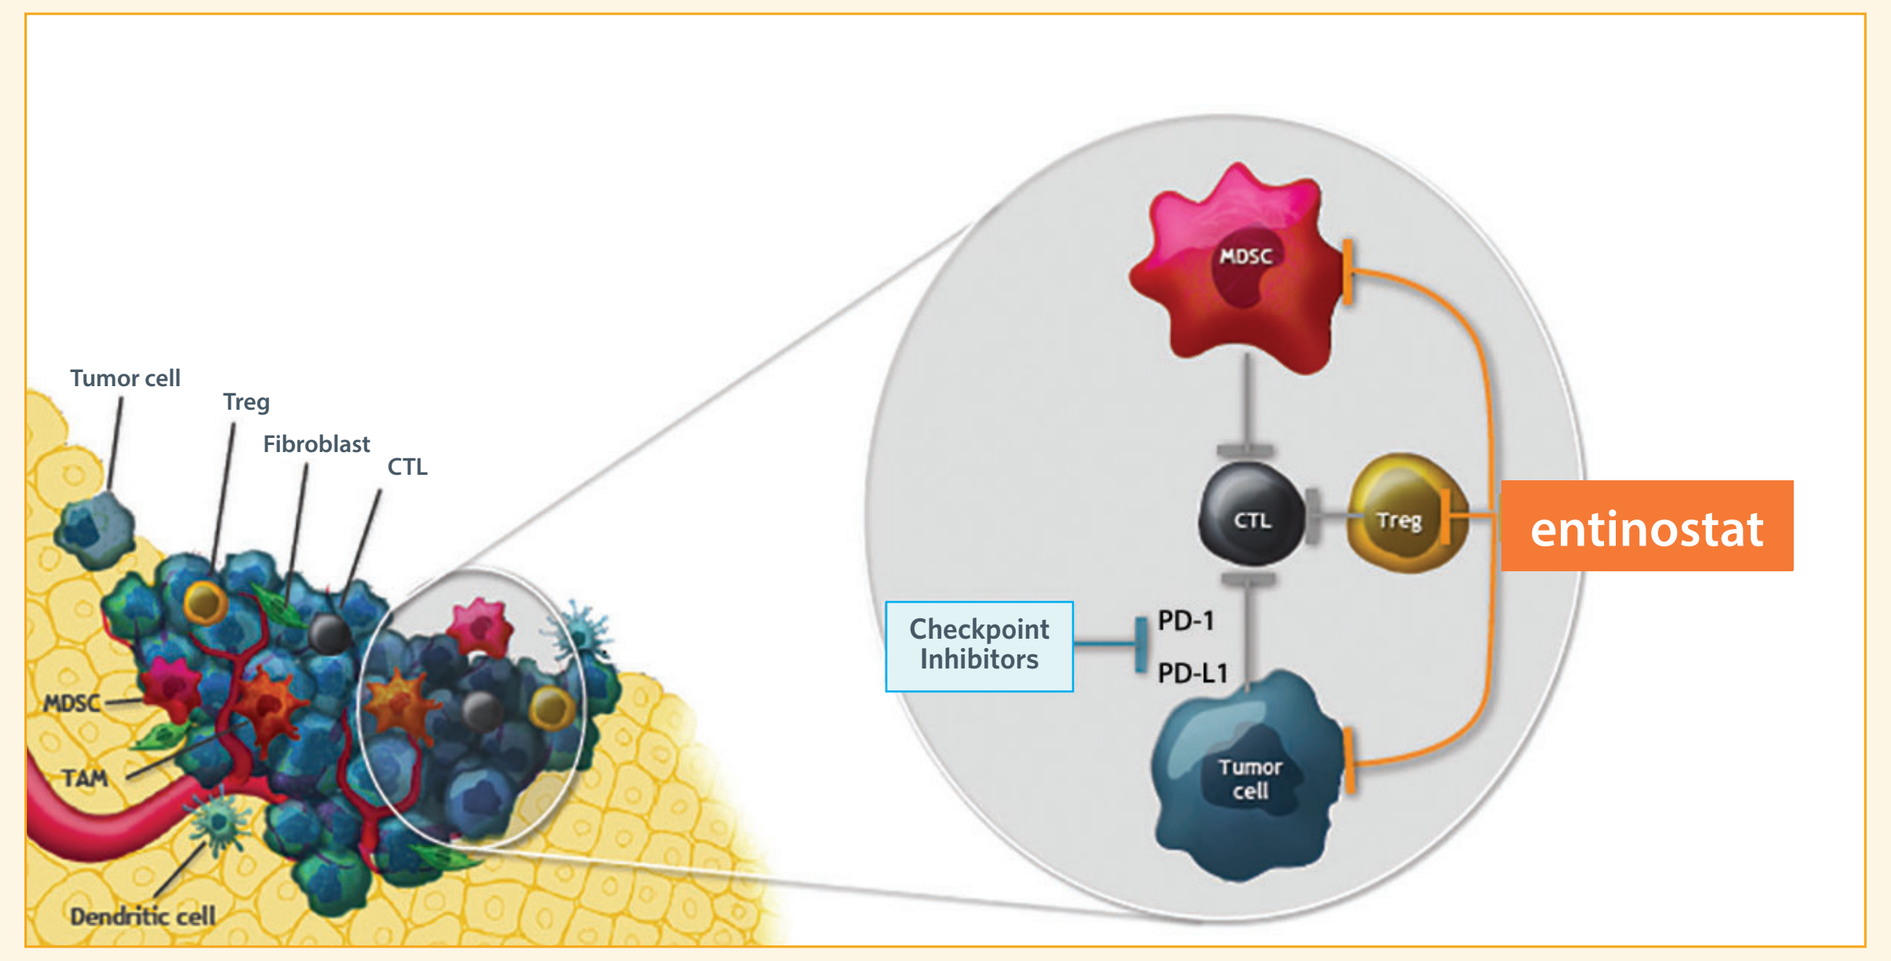
Tumor cell
Treg
Fibroblast
CTL
MDSC
TAM
Dendritic cell
MDSC
CTL
Treg
entinostat
Checkpoint Inhibitors
PD-1
PD-L1
Tumor cell

CTL, cytotoxic T lymphocyte; MDSC, myeloid derived suppressor cell; TAM, tumor-associated macrophage; Treg, regulatory T lymphocyte.

METHODS

Key Patient Eligibility Criteria

- Histologically- or pathologically-confirmed recurrent or metastatic NSCLC and previously treated with and unequivocally progressed on either a PD-1 or PD-L1-blocking antibody.
- Previously treated with at least 1 chemotherapeutic regimen in the advanced/metastatic setting, and if patient has EGFR mutation-positive or ALK translocation-positive disease, must have received an EGFR inhibitor or ALK inhibitor.
- Patients were eligible regardless of histology or baseline PD-L1 expression.
- Other eligibility criteria included ECOG Performance Status 0 or 1, measurable disease per RECIST 1.1, and willingness to have fresh tumor samples collected during screening and at other time points as necessary.

Study Design

The Phase 2 expansion phase of ENCORE 601 utilizes a Simon 2-stage design to assess activity. The number of patients evaluated in each stage of this cohort was based on a single proportion binomial test with 90% power and a 1-sided significance level of 5%. Three responses in 31 patients were observed in Stage 1 of the study, meeting the criteria to expand to Stage 2 and enroll up to 56 patients[5]. The study was further revised to accrue up to 70 patients to increase statistical power and decrease Type I error.

The primary endpoint is overall response rate (ORR) as assessed by irRECIST.

METHODS (continued)

Higher levels of circulating classical monocytes ($CD14^+CD16^-HLA\text{-}DR^{hi}$) have recently been shown to associate with improved clinical outcome to anti-PD-1 blockade in melanoma patients[6]. The role of circulating classical monocytes and their representation of the tumor microenvironment in anti-PD-1 relapsed/refractory NSCLC has not been reported. In blood samples obtained at baseline in this clinical trial, we measured circulating classical monocytes to determine whether there was an association of monocyte levels with clinical benefit to the entinostat + pembrolizumab combination. In addition, we analyzed gene expression of fresh tumor biopsies obtained prior to treatment to evaluate whether circulating monocyte levels were representative of the immune state of the tumor microenvironment.

Treatment Assessments

- All patients received entinostat 5 mg QW PO + pembrolizumab 200 mg Q3W IV in 21-day cycles until disease progression or discontinuation for other reasons.
- Disease assessments were performed every 6 weeks.
- Classical peripheral blood monocytes were characterized as CD14-positive, CD16-negative, and HLA-DR high. Classical monocyte data were available for 51 of 57 patients.

PATIENT DEMOGRAPHICS

Demographics

- First patient was enrolled December 2015, and last patient was enrolled September 2017. Of the first 57 patients enrolled, 22 had refractory disease to prior PD-(L)1 therapy, and 4 had a documented prior response.
- Median duration of prior PD-(L)1 therapy was < 6 months and the median time from last dose of prior PD-(L)1 therapy was 65 days.
- Additional baseline characteristics are noted in **Table 1.**

Table 1: Demographics	N=57
Gender, n (%)	
Female	24 (42.1)
Male	33 (57.9)
Age, (years)	
Median	66.0
Range	48 to 85
Race, n (%)	
White	49 (86.0)
Black or African American	3 (5.3)
Other (Specify)	5 (8.8)
ECOG PS, n (%)	
Grade 0 = Normal activity	14 (24.6)
Grade 1 = Symptoms, but ambulatory	42 (73.7)
Missing	1 (1.8)
Smoking Status, n (%)	
Current Smoker	2 (3.5)
Former Smoker	50 (87.7)
Never Smoker	5 (8.8)
PD-L1 Expression, n (%)*	
<1%	21 (36.8)
1-49%	20 (35.1)
>= 50%	8 (14.0)
Not Evaluable	8 (14.0)
Stage of Disease, n (%)	
IIIA	5 (8.8)
IIIB	2 (3.5)
IV	48 (84.2)
Missing	2 (3.5)
Visceral Involvement, n (%)	
No	10 (17.5)
Yes	45 (78.9)
Unknown	2 (3.5)
LDH (>ULN), n (%)	
No	38 (66.7)
Yes	18 (31.6)
Unknown	1 (1.8)
Lines of Prior Therapy, median	3
PD1 Antagonist as Immediate Prior Therapy, n (%)	38 (66.7)
Best Response on Prior PD-1/PD-L1 Therapy, n (%)	
Complete Response	1 (1.8)
Partial Response	3 (5.3)
Stable Disease	27 (47.4)
Disease Progression	22 (38.6)
Unknown	4 (7.0)
Duration on Prior PD-1/PD-L1 Therapy, (Days)	
Median	162.0
Range	19 to 693
Duration Between Last Dose of Prior PD-1/PD-L1 therapy	
Median	65.0
Range	21 to 1614

*Fresh biopsy obtained at screening.
ECOG, Eastern Cooperative Oncology Group; LDH, lactate dehydrogenase; PS, performance status; ULN, upper limit of normal.

EFFICACY

- **Figure 2** shows that 6 patients out of 57 had a confirmed PR, resulting in a 11% ORR (95% CI: 4-21%).
 - 4 responders had baseline PD-L1 expression <1% as shown in **Table 2**.
- The median duration of response is 4.5 months, with the longest ongoing over 14 months, as shown in **Figure 3**.
- Median progression-free survival was 82 days (95% CI: 43, 124).
- Stable disease was observed in 25/57 (44%) patients.
- Clinical benefit rate, defined as complete or partial response or stable disease for 24 weeks or more from initiation of treatment, was 18% (10/57 patients). The change in tumor size for these patients can be seen in **Figure 4**.
- At the data cutoff, 7 patients remain ongoing, including 4/6 partial responders. **(Figure 3 and Table 2)**

Figure 2. **Percentage Change in Tumor Size from Baseline (Sum of Diameters of Target Lesions).** Six confirmed partial responses have been observed to date with an ORR of 11%.

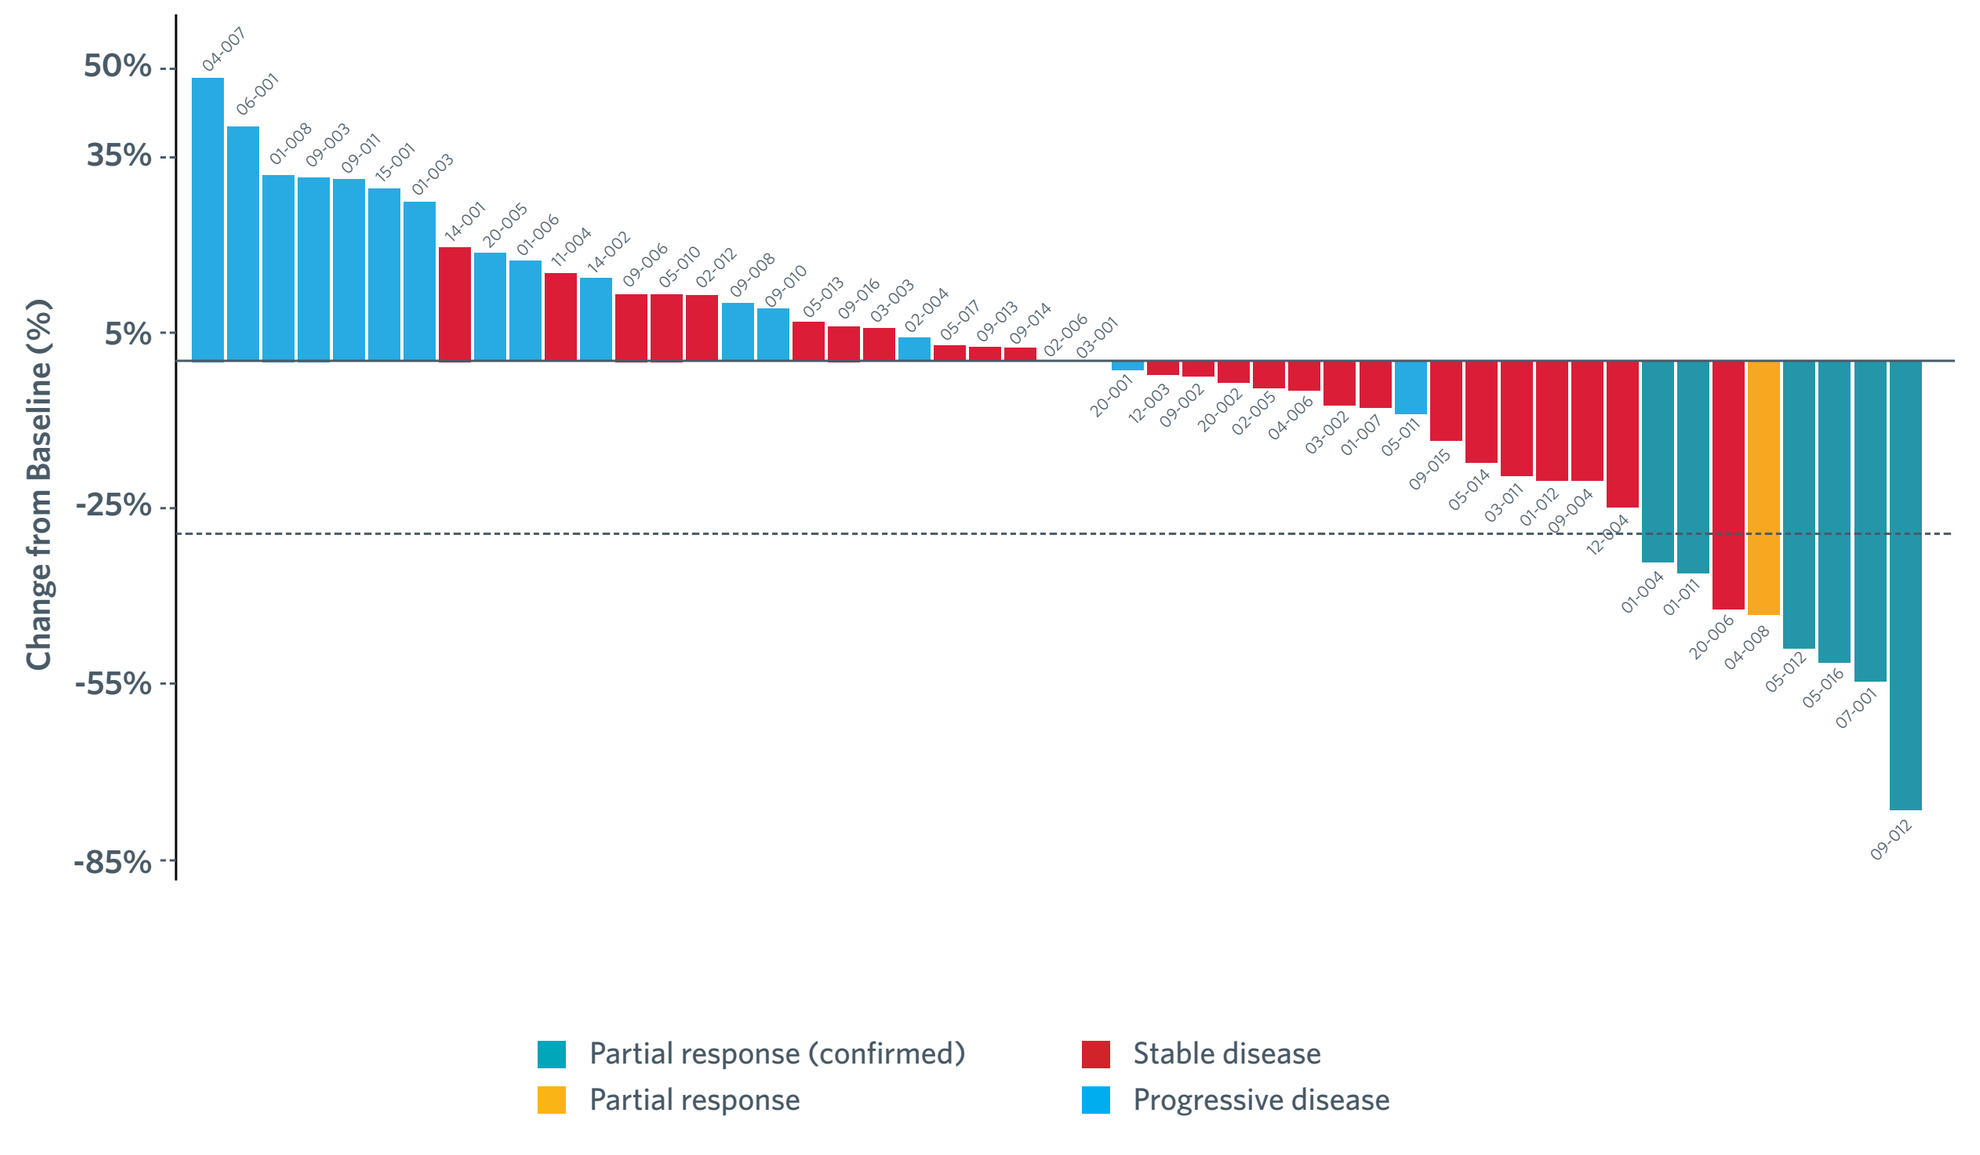
Change from Baseline (%)
50%
35%
5%
-25%
-55%
-85%
Partial response (confirmed)
Partial response
Stable disease
Progressive disease

Figure 3. **Patient Response and Time on Treatment.** irRECIST patient response by investigator assessment. **Red** patient numbers represent those with high baseline classical monocyte levels. Median duration of response was 4.5 months.

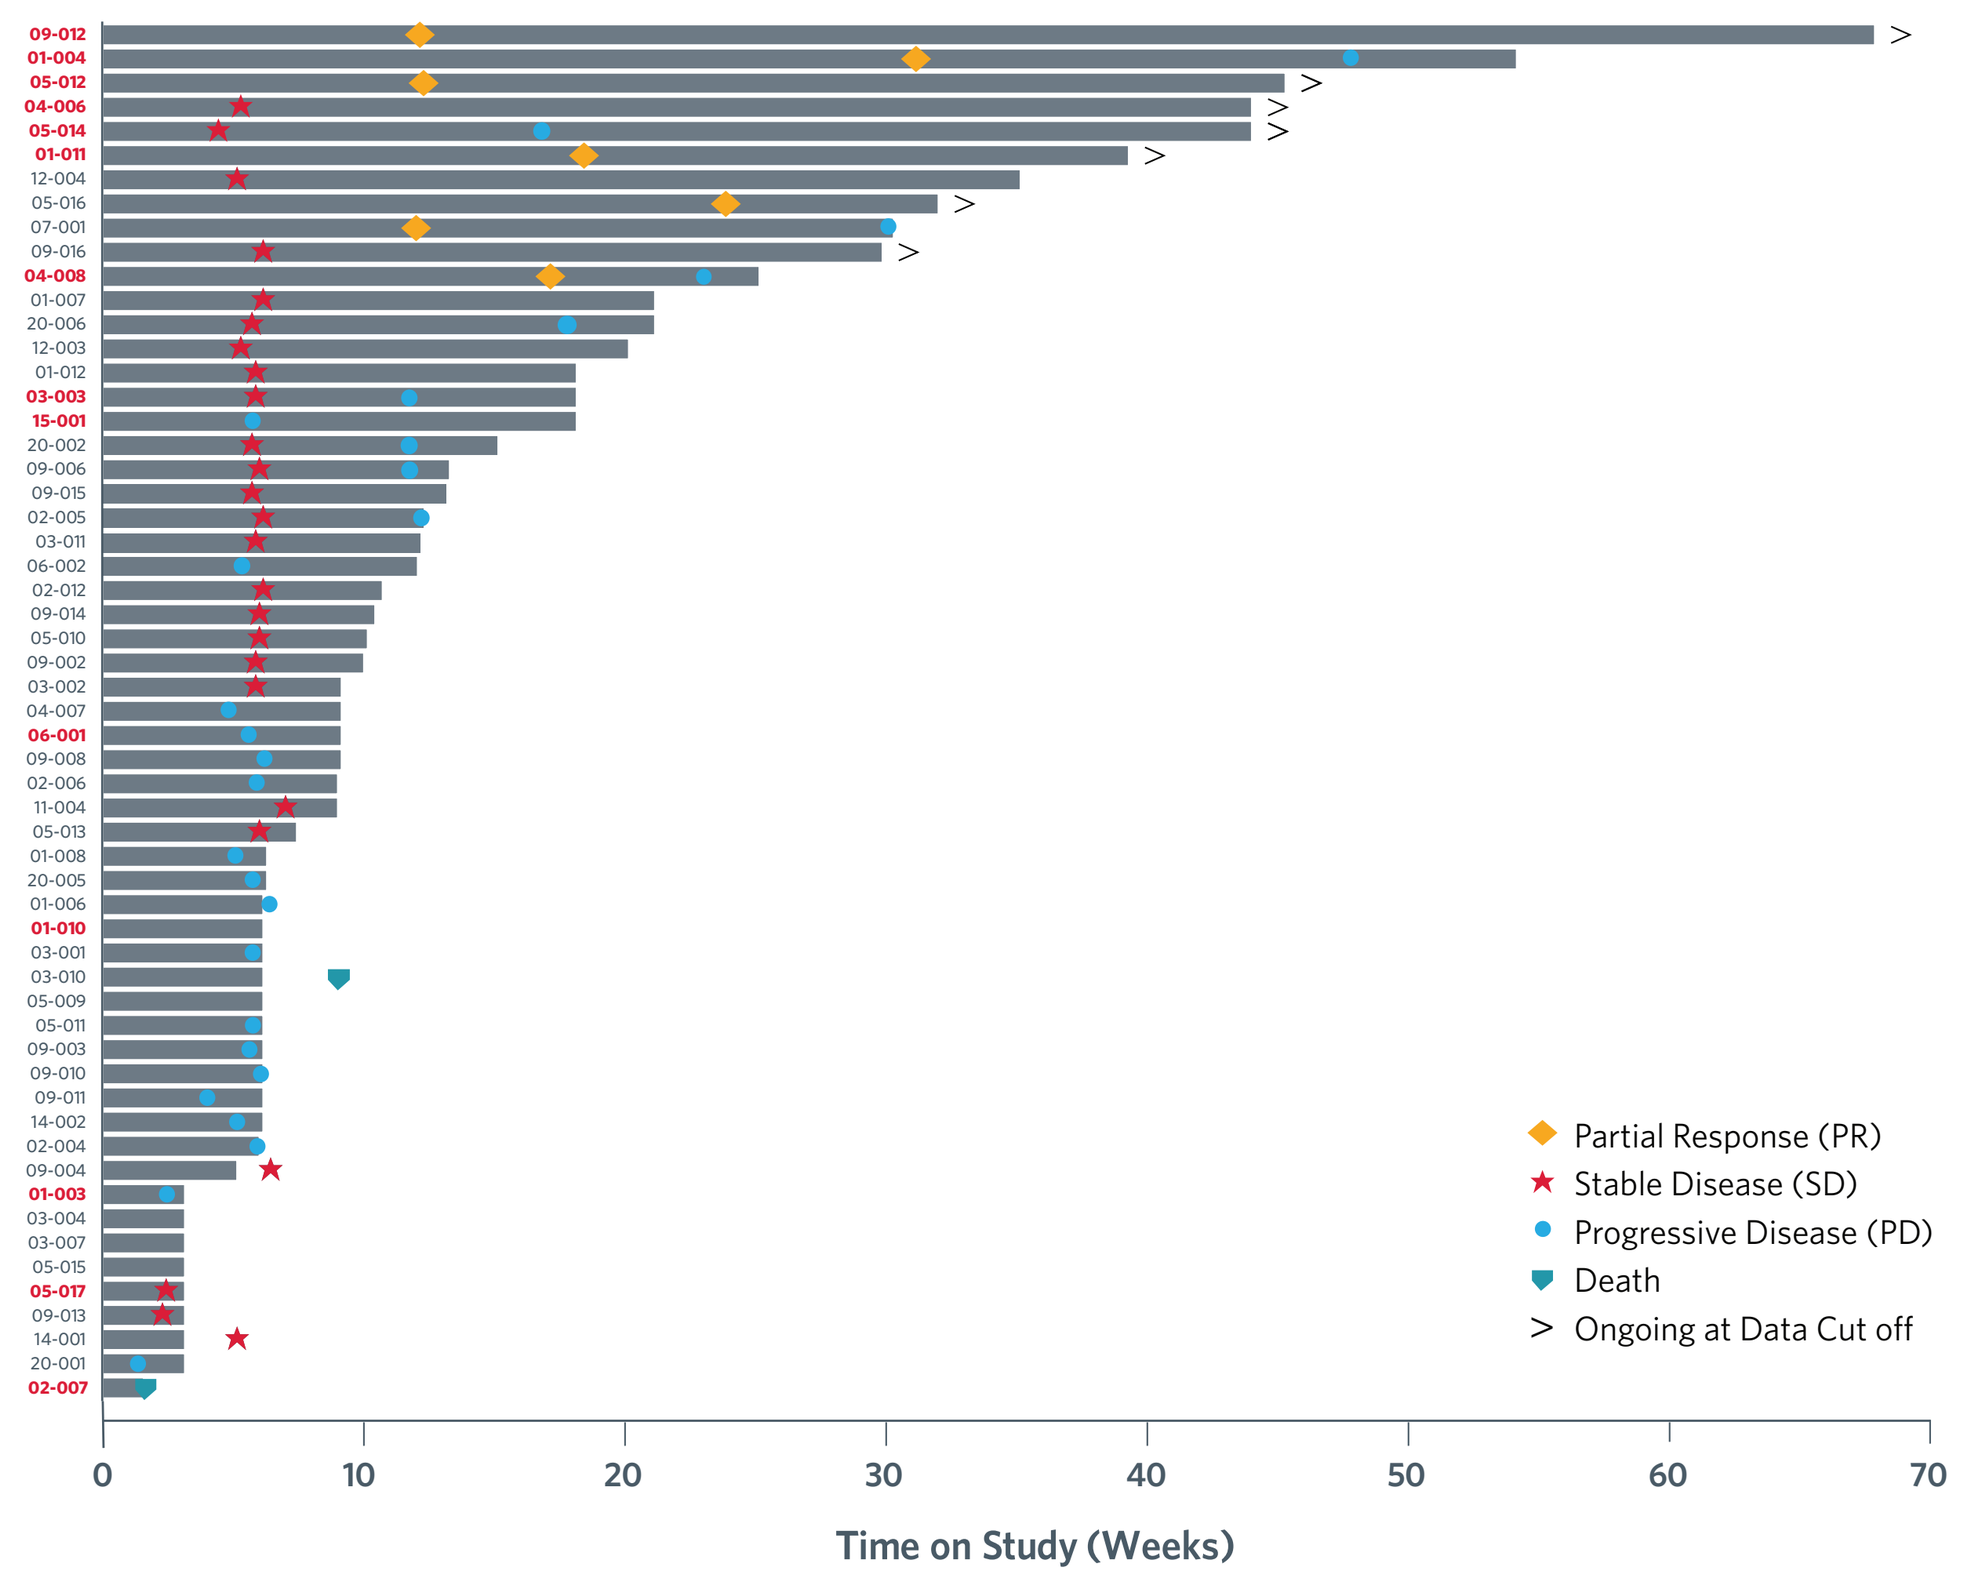
Time on Study (Weeks)
0
10
20
30
40
50
60
70
Partial Response (PR)
Stable Disease (SD)
Progressive Disease (PD)
Death
> Ongoing at Data Cut off

EFFICACY (continued)

Figure 4. **Change in Tumor Size from Baseline Over Time.**

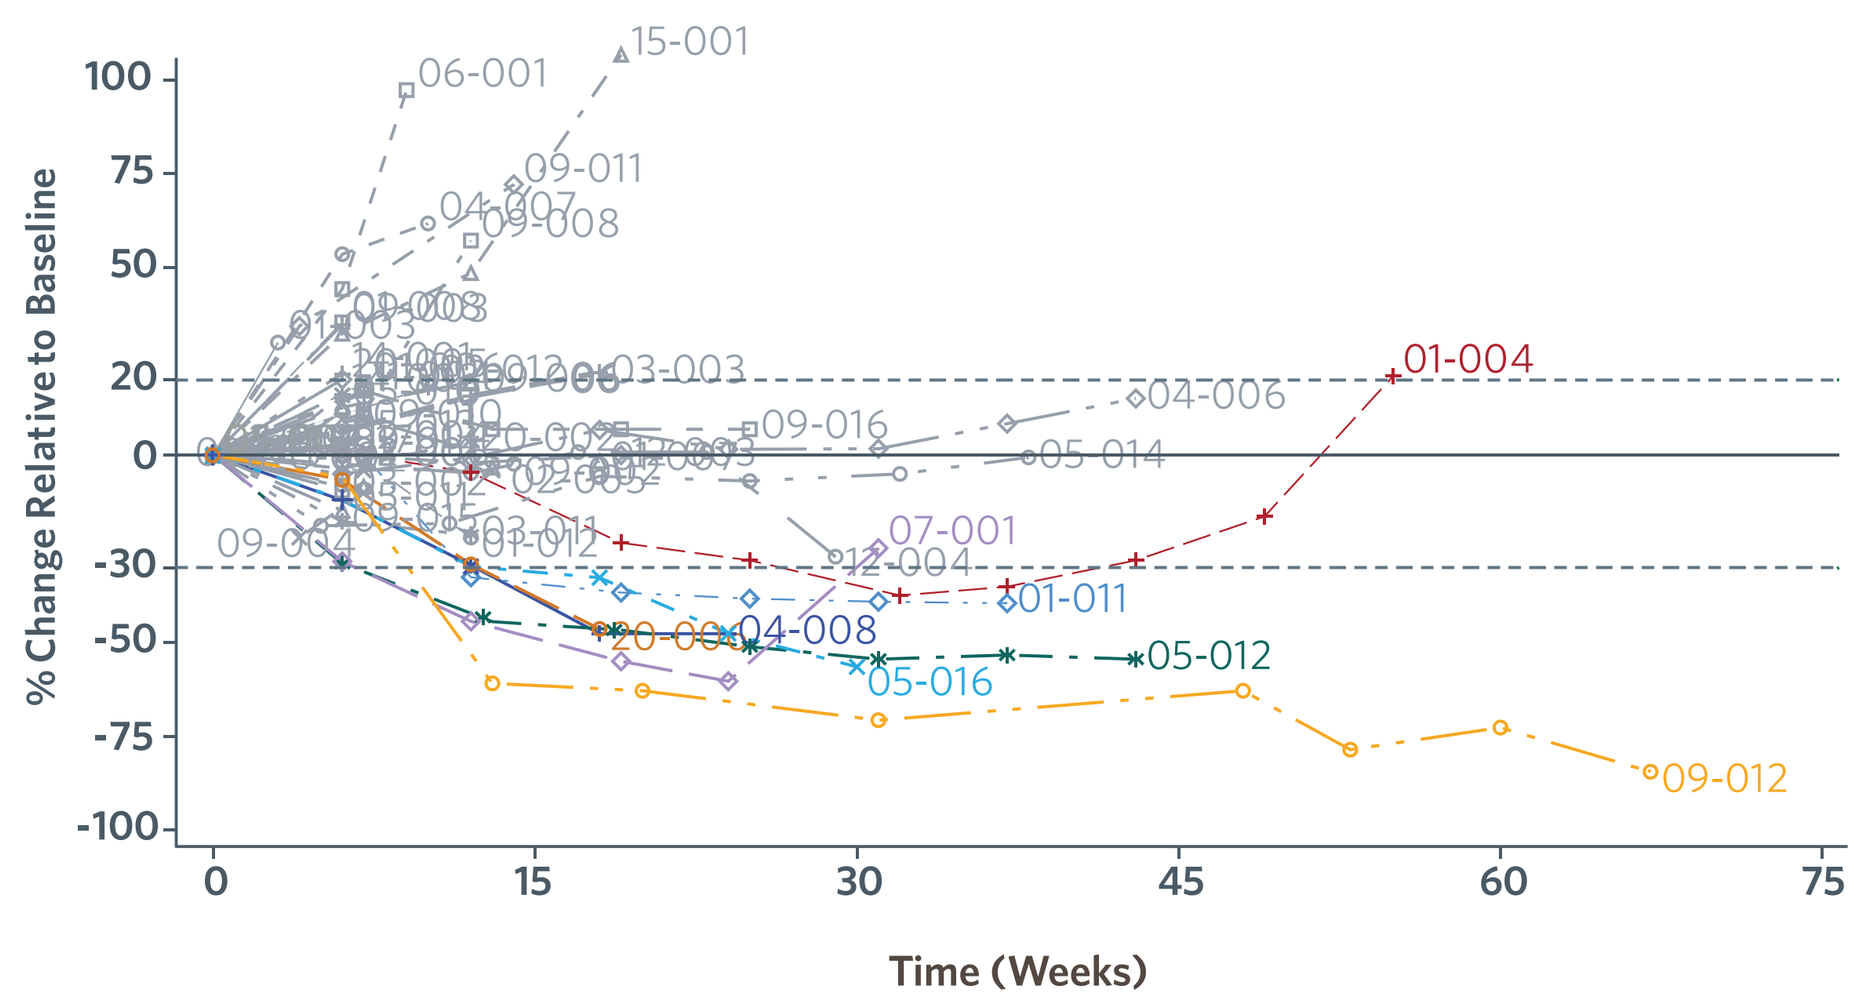
% Change Relative to Baseline
Time (Weeks)
100
75
50
20
0
-30
-50
-75
-100
0
15
30
45
60
75
15-001
06-001
09-011
09-008
01-004
04-006
05-014
09-016
07-001
01-011
04-008
05-012
05-016
09-012

Table 2. **Characteristics of Patients with Confirmed Partial Response.** Six confirmed partial responses have been observed, the majority were PD-L1 negative by fresh biopsy. Four of the six patients are ongoing in the study.

Patient	Baseline PD-L1 Expression	EGFR/ALK Testing	Prior Anti-PD-1	Best Response to Prior Anti-PD-1 (mo)	Duration on Prior Tx (mo)	Time Since Last Dose Prior Anti-PD-1 (mo)	Duration of Response on Trial (mo)	Status at Time of Data Cutoff
09-012	1-49%	- / -	Pembrolizumab	SD	12.8	1.1	12.9+	Ongoing
05-012	<1%	- / -	Nivolumab	PR	9.7	10.8	7.6+	Ongoing
01-011	<1%	NA / NA	Nivolumab	SD	5.2	1.4	4.8+	Ongoing
07-001	<1%	- / NA	Pembrolizumab	SD	19.7	17.3	4.2	Discontinued due to PD
01-004	<1%	NA / NA	Nivolumab	Unknown	7.3	1.7	3.9	Discontinued due to PD
05-016	≥50%	- / -	Nivolumab	PR	22.8	0.7	1.9+	Ongoing

CORRELATES

- Classical monocytes were obtained from peripheral blood and were defined as CD14+, CD16-, and HLA-DR high.
- **Figure 5** shows that patients with clinical benefit from treatment with entinostat and pembrolizumab had higher baseline classical monocytes than those without response or healthy donors. Patients with high classical monocytes and their response to therapy can be seen in red in **Figure 3**.
- Progression-free survival (5.4 months) and overall response rate (29%) to entinostat + pembrolizumab was improved for patients with elevated classical monocytes compared to those with lower classical monocytes **(Figure 6)**.

Figure 5. **Higher Baseline Classical Monocyte Levels were Associated with Clinical Benefit to ENT/PEMBRO and an Inflamed Gene Expression Profile.** Classical peripheral blood monocytes (CD14+,CD16-,HLA-DR[hi]) were available for 51 of 57 patients.

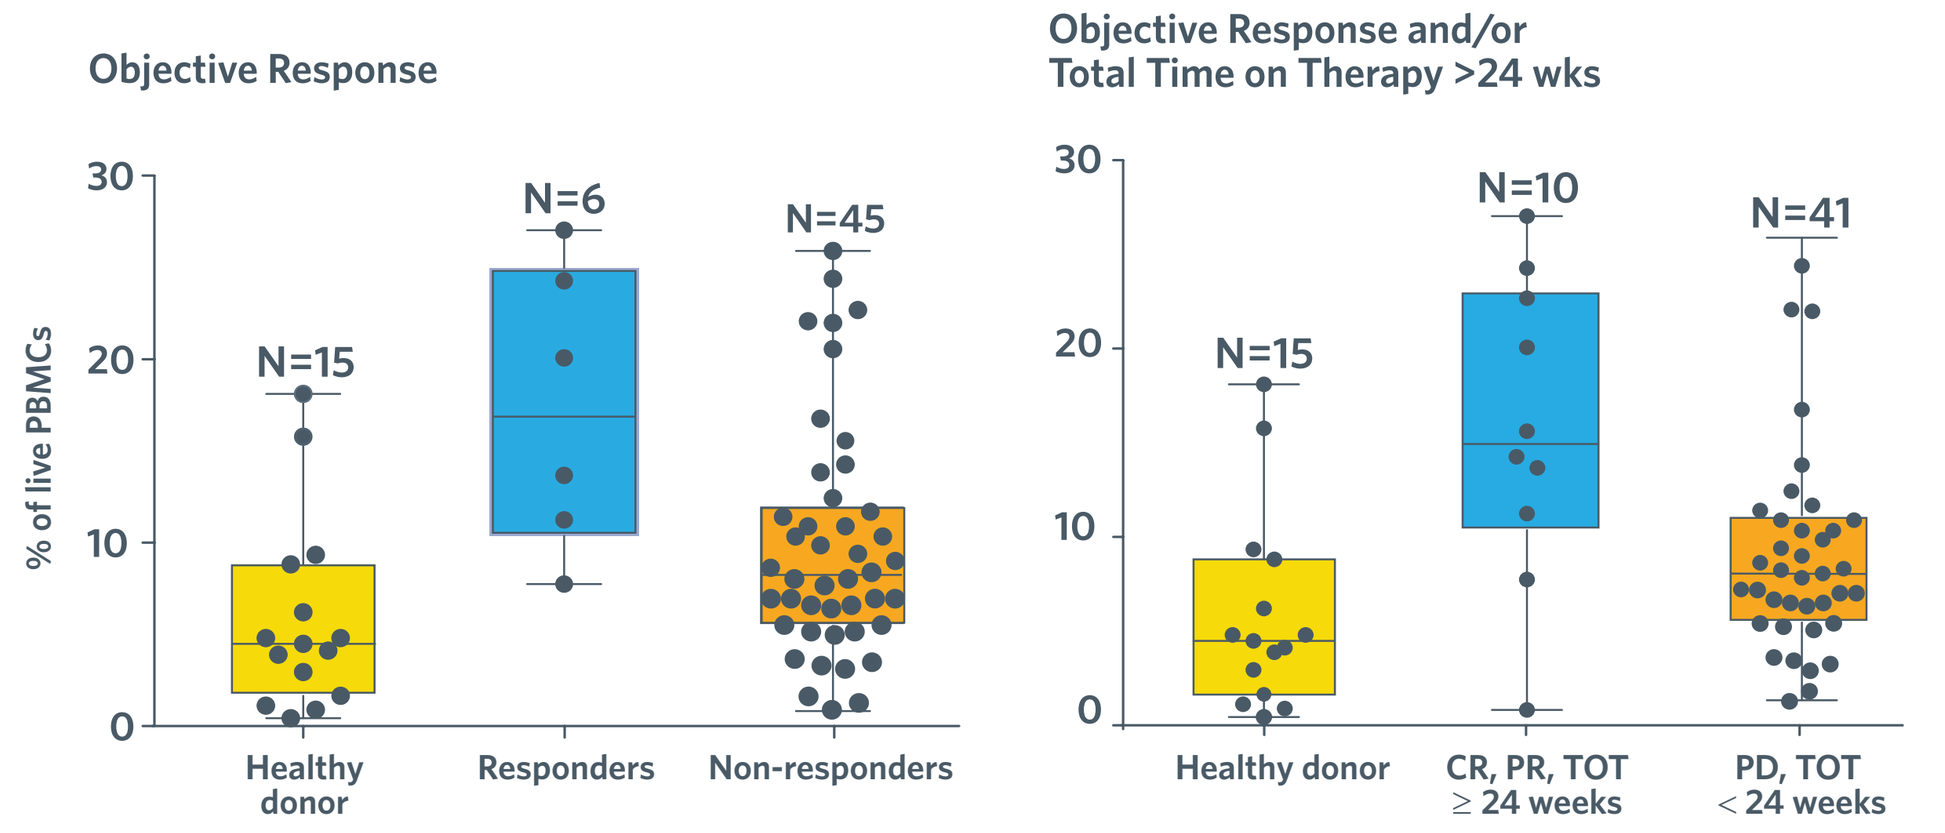
Objective Response
Objective Response and/or Total Time on Therapy >24 wks
% of live PBMCs
30
20
10
0
N=15
N=6
N=45
N=15
N=10
N=41
Healthy donor
Responders
Non-responders
Healthy donor
CR, PR, TOT ≥ 24 weeks
PD, TOT < 24 weeks

CR, complete response; PD, progressive disease; PR, partial response; TOT, time on treatment.

CORRELATES (continued)

Figure 6. **Higher Baseline Levels of Classical Monocytes are Associated with PFS and ORR Benefits.** Among patients with elevated classical monocytes, ORR was 28.6% and median PFS was 5.4 months.

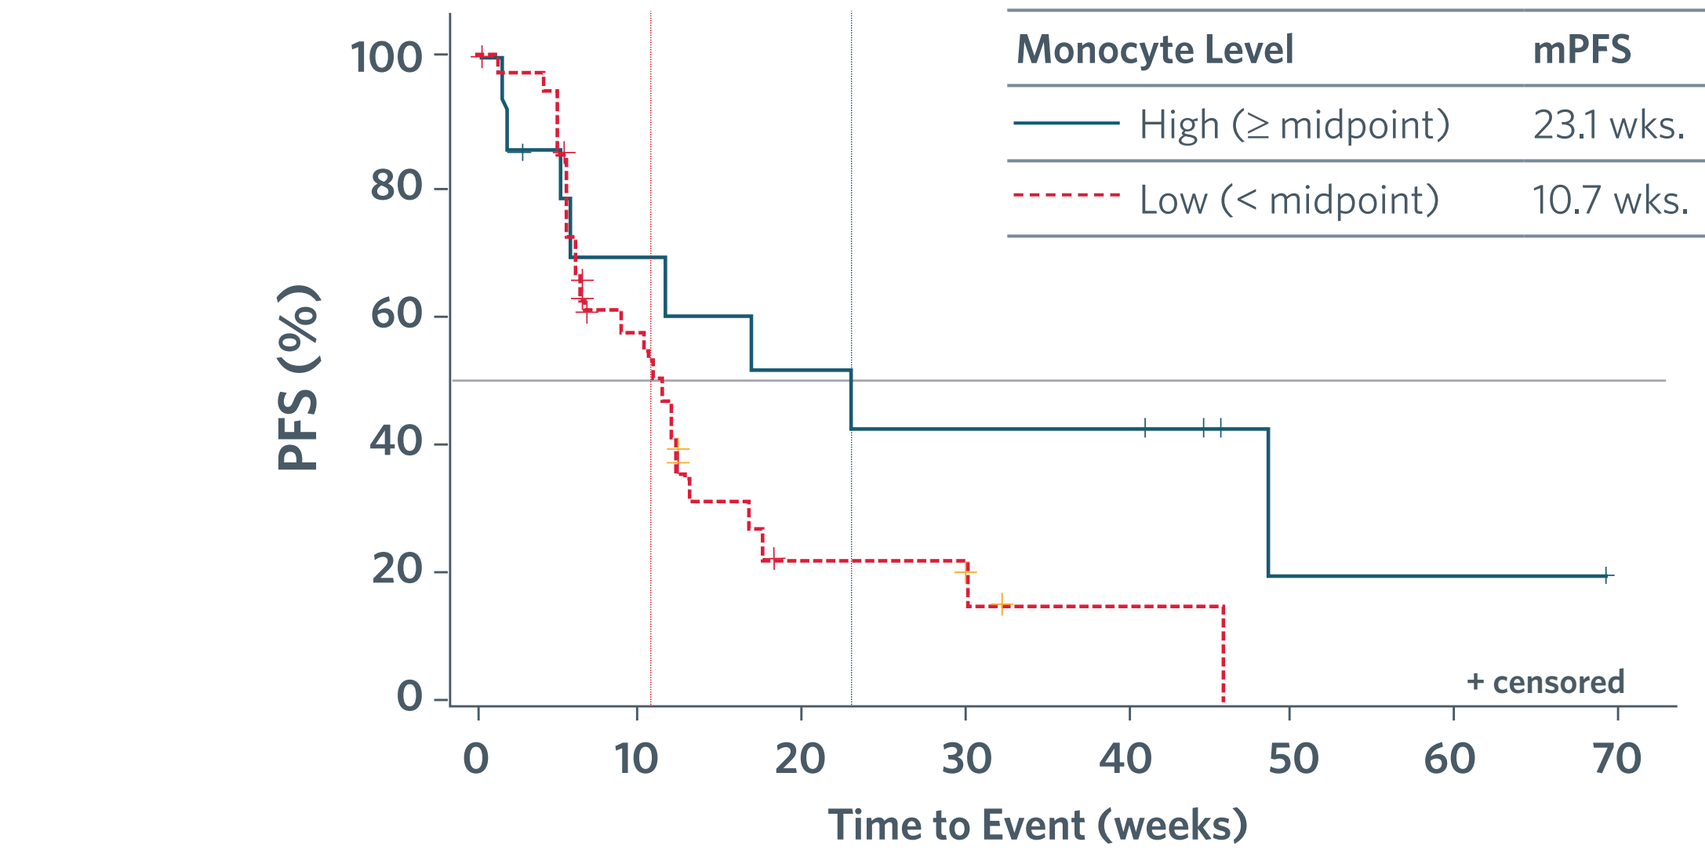
PFS (%)
100
80
60
40
20
0
0
10
20
30
40
50
60
70
Time to Event (weeks)
+ censored
Monocyte Level
mPFS
ORR
High (≥ midpoint)
23.1 wks.
28.6%
Low (< midpoint)
10.7 wks.
5.4%

Monocyte High	14	8	6	5	4	1	1	0
Monocyte Low	37	16	4	3	1	0		

Figure 7. **Upregulation of Tumor Inflammation Pathways in Patients with Elevated Monocytes.** Nanostring PanCancer IO 360TM panel assessed in samples with high baseline monocytes and gene expression was compared to that in low monocyte samples.

A)

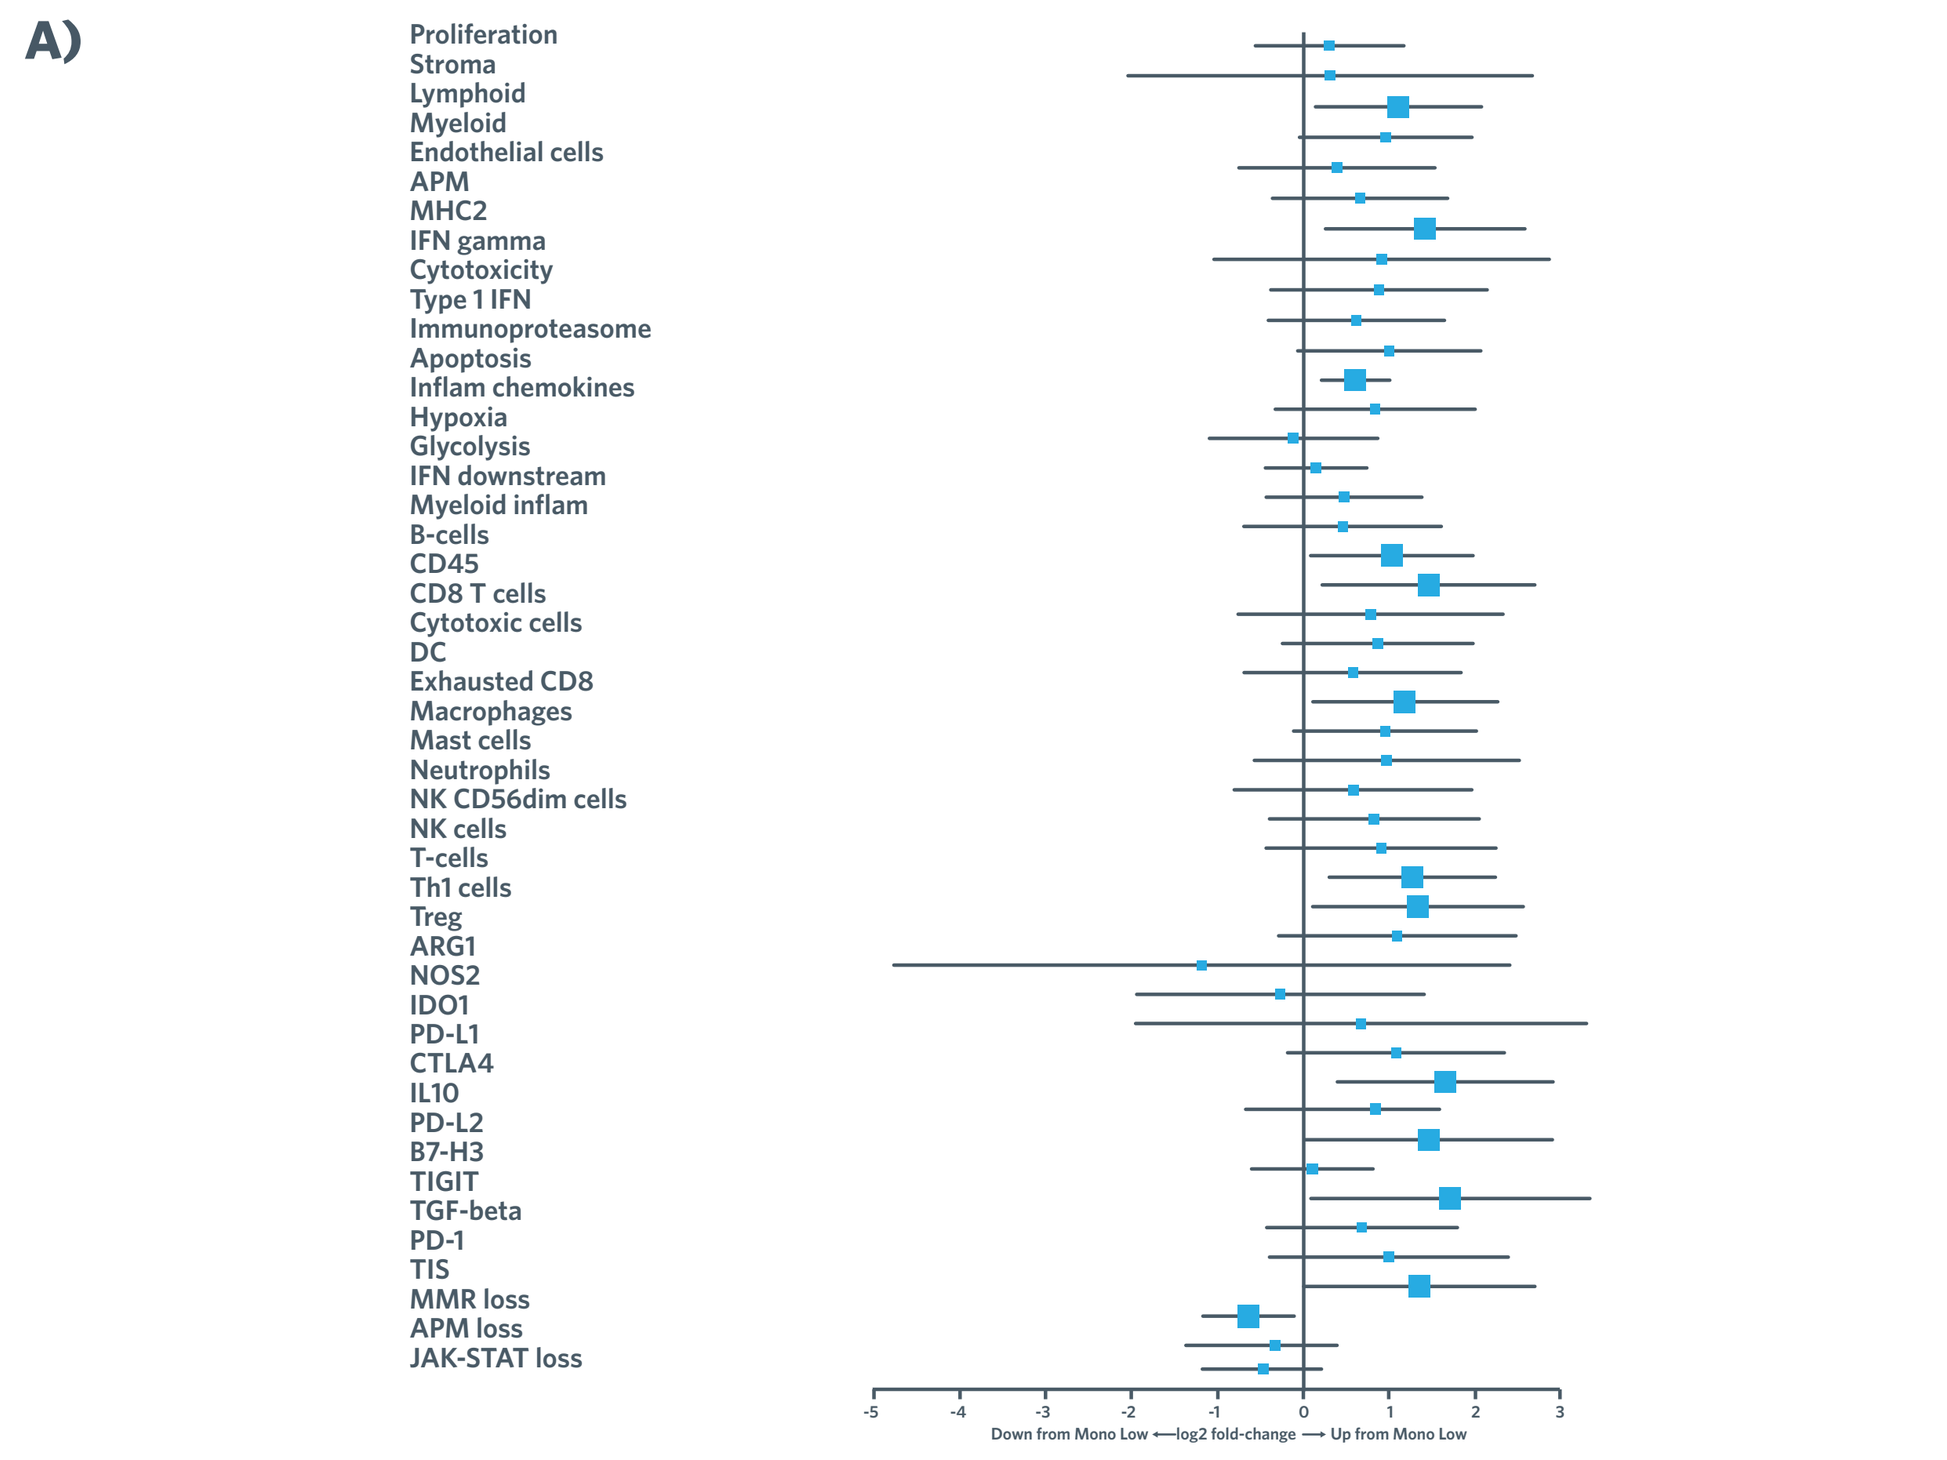
Proliferation
Stroma
Lymphoid
Myeloid
Endothelial cells
APM
MHC2
IFN gamma
Cytotoxicity
Type 1 IFN
Immunoproteasome
Apoptosis
Inflam chemokines
Hypoxia
Glycolysis
IFN downstream
Myeloid inflam
B-cells
CD45
CD8 T cells
Cytotoxic cells
DC
Exhausted CD8
Macrophages
Mast cells
Neutrophils
NK CD56dim cells
NK cells
T-cells
Th1 cells
Treg
ARG1
NOS2
IDO1
PD-L1
CTLA4
IL10
PD-L2
B7-H3
TIGIT
TGF-beta
PD-1
TIS
MMR loss
APM loss
JAK-STAT loss
Down from Mono Low ⟵log2 fold-change⟶ Up from Mono Low

B)

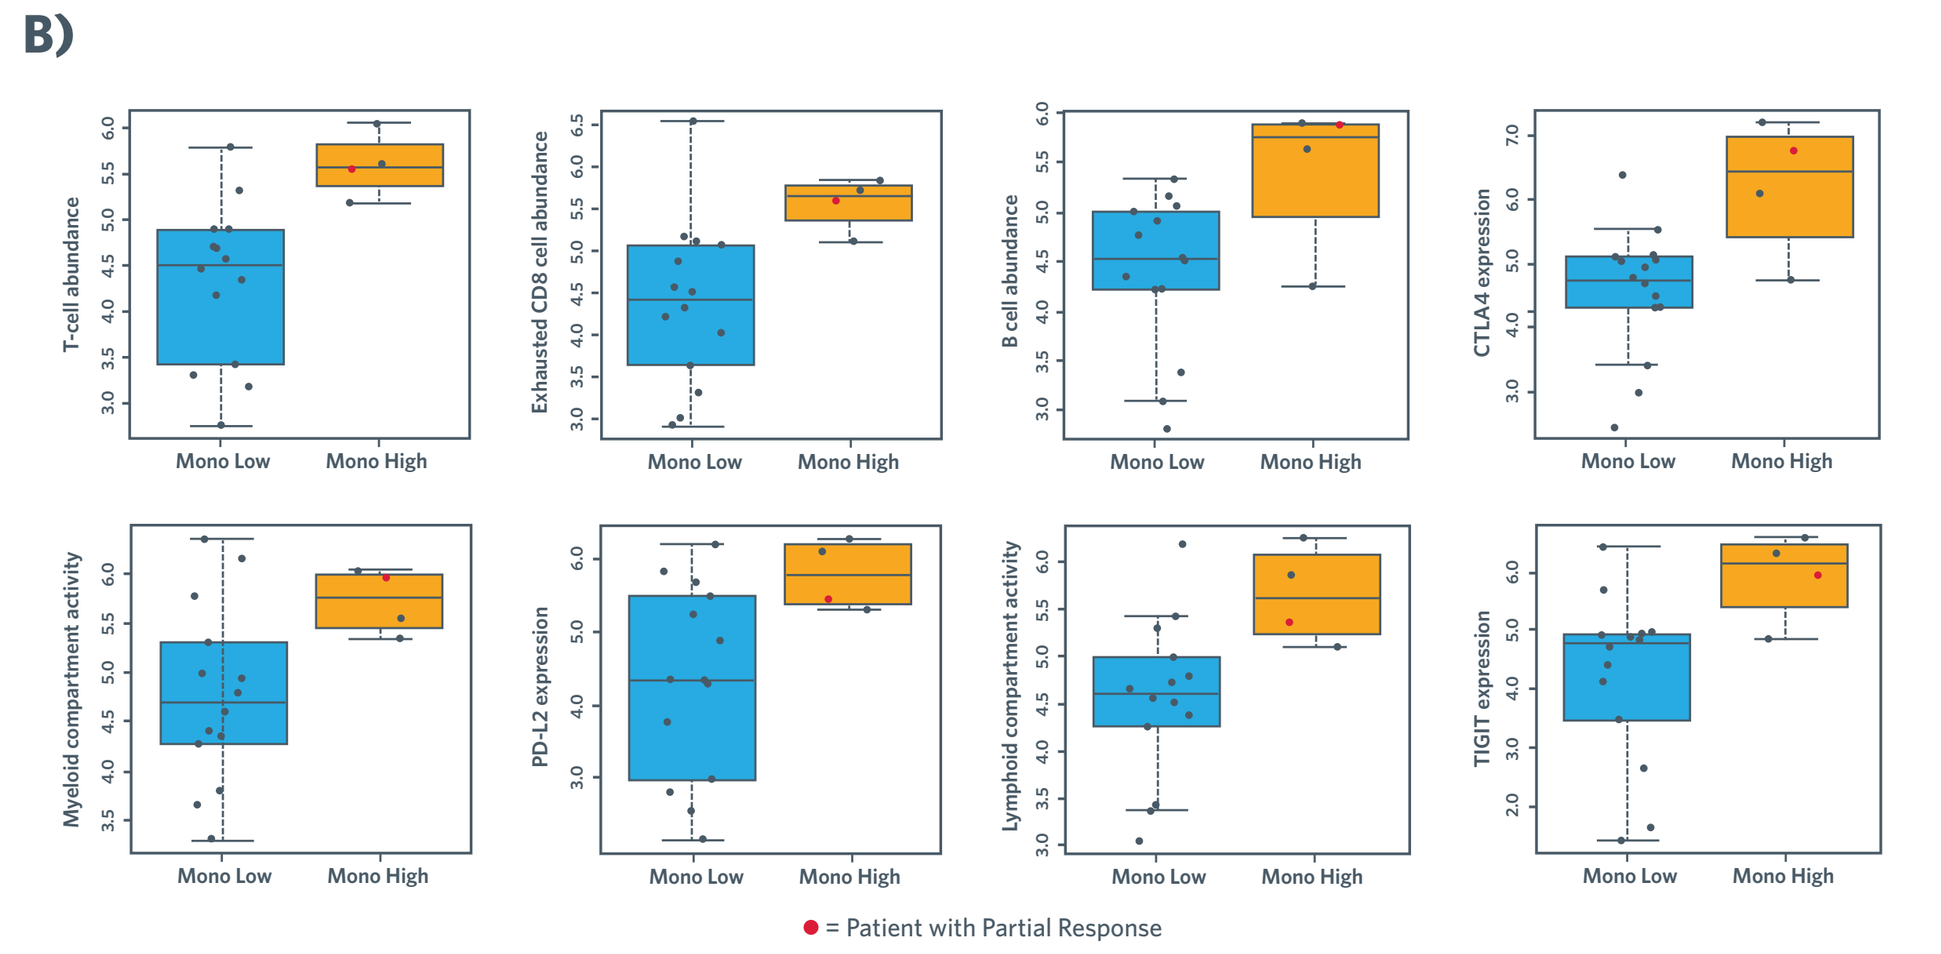
T-cell abundance
Exhausted CD8 cell abundance
B cell abundance
CTLA4 expression
Myeloid compartment activity
PD-L2 expression
Lymphoid compartment activity
TIGIT expression
Mono Low
Mono High
= Patient with Partial Response

- Tumor biopsy samples were obtained at screening and gene expression was assessed on the NanoString PanCancer IO 360™ panel. On C1D1, relative blood monocytes levels were assessed by flow cytometry.
- Signature analysis reveals that several pathways associated with tumor inflammation are upregulated in samples from patients with elevated monocyte levels **(Figure 7A)**.
- Signatures specific to increased immune function were observed **(Figure 7B)**, as well as signatures consistent with immune exhaustion in patients resistant to checkpoint inhibition.

SAFETY

- **Table 3** shows treatment-emergent adverse events related to study treatment (entinostat or pembrolizumab or both) observed in >10% of patients at any grade, or in any patients at grade ≥3.
- TEAEs Grade 3 or higher related to entinostat or pembrolizumab or both occurred in 24/57 (42.1%) patients.
 - 5 pts (8.8%) experienced Grade 3/4 related irAEs (2 events each of pneumonitis and colitis, 1 event of hyperthyroidism). In addition, 19 pts (33.3%) experienced other Grade 3/4 related AEs with only fatigue, anemia, hypophosphatemia, and hyponatremia occurring in more than 2 patients.
- Treatment-related AEs led to discontinuation in 12 patients (22.2%). These included fatigue (3), pneumonitis (2), encephalitis, acute respiratory failure, hyponatremia, ventricular arrhythmia, asthenia, colitis, and vomiting/diarrhea.

Table 4: Safety	Any Grade (N=57)	Grade ≥3 (N=57)
Subjects With At Least One Treatment-Emergent Adverse Event Related to Study Treatment	44 (77.2)	24 (42.1)
Gastrointestinal disorders		
Nausea	7 (12.3)	
Diarrhea	11 (19.3)	2 (3.5)
Vomiting	6 (10.5)	1 (1.8)
Colitis	2 (3.5)	2 (3.5)
Abdominal pain	1 (1.8)	1 (1.8)
General disorders and administration site conditions		
Fatigue	23 (40.4)	5 (8.8)
Asthenia	1 (1.8)	1 (1.8)
Respiratory, thoracic and mediastinal disorders		
Pneumonitis	5 (8.8)	2 (3.5)
Pleural effusion	2 (3.5)	1 (1.8)
Acute respiratory failure	1 (1.8)	1 (1.8)
Investigations		
Platelet count decreased	10 (17.5)	1 (1.8)
Weight decreased	6 (10.5)	
Metabolism and nutrition disorders		
Decreased appetite	12 (21.1)	
Hypophosphatemia	6 (10.5)	4 (7.0)
Hyponatremia	6 (10.5)	3 (5.3)
Hyperglycemia	1 (1.8)	1 (1.8)
Hypokalemia	1 (1.8)	1 (1.8)
Cardiac disorders		
Atrial fibrillation	1 (1.8)	1 (1.8)
Ventricular arrhythmia	1 (1.8)	1 (1.8)
Blood and lymphatic system disorders		
Anemia	11 (19.3)	4 (7.0)
Infections and infestations		
Clostridium difficile colitis	1 (1.8)	1 (1.8)
Encephalitis	1 (1.8)	1 (1.8)
Psychiatric disorders		
Mental status changes	1 (1.8)	1 (1.8)
Endocrine disorders		
Hyperthyroidism	1 (1.8)	1 (1.8)

CONCLUSIONS

- ENT + PEMBRO demonstrated anti-tumor activity and acceptable safety in patients with NSCLC who have progressed on prior PD-(L)1 blockade.
- Preliminary results indicate that higher levels of circulating classical monocytes are associated with clinical benefit to the ENT + PEMBRO combination in this anti-PD-1 and chemotherapy relapsed/refractory NSCLC patient cohort.
- Further, our data indicate that higher levels of classical monocytes may correlate with an inflamed tumor microenvironment that is poised to respond to anti-PD1 blockade with the addition of entinostat to relieve immunosuppression and restore a robust anti-tumor T cell response.
- Measurement of circulating levels of classical monocytes may serve as a biomarker to be validated prospectively to select for patients who will benefit from entinostat + anti-PD1 treatment.

References:

1. Zimmer L, et al. *Eur J Cancer.* 2017;75:47-55.
2. Orillion A, et al. *Clin Cancer Res.* 2017;23:5187-5201.
3. Johnson ML, et al. Presented at the ASCO Annual Meeting, 2017. Abstract 9529.
4. Johnson ML, et al. Presented at SITC Annual Meeting, 2017. Abstract P72.
5. Gandhi L, et al. Presented at SITC Annual Meeting, 2017. Abstract O19.
6. Krieg C, et al. *Nature Medicine.* 2018;24:144-153.

Acknowledgements:

The authors thank the patients and their families, investigators and study staff. We acknowledge NanoString® Technologies, Inc. for providing the PanCancer IO 360™ panels for this work. We also thank Amy Sullivan, Sarah Church, Elena Viboch of the Nanostring Team for their contributions to this analysis and our ongoing collaboration. This study was sponsored by Syndax Pharmaceuticals, Inc., in collaboration with Merck & Co., Inc., Kenilworth, NJ. Writing and editorial assistance was provided by BluPrint Oncology, LLC. This assistance was funded by Syndax Pharmaceuticals, Inc.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Poster presented at the American Society of Clinical Oncology Annual Meeting, June 1-5, 2018, Chicago IL.