O19

ENCORE-601: Phase 1b/2 Study of Entinostat (ENT) in Combination With Pembrolizumab (PEMBRO) in Patients With Non-Small Cell Lung Cancer

Leena Gandhi,[1] Melissa L. Johnson,[2] Mateusz Opyrchal,[3] Suresh Ramalingam,[4] Pasi A. Jänne,[5] Abraham Chachoua,[1] Peter Ordentlich,[6] Susan Brouwer,[6] Serap Sankoh,[6] Emmett Schmidt,[7] Michael L. Meyers,[8] Matthew D. Hellmann[9]

[1]NYU Langone Medical Center, New York, NY; [2]Sarah Cannon Research Institute, Nashville, TN; [3]Roswell Park Cancer Institute, Buffalo, NY; [4]Winship Cancer Institute, Emory University, Atlanta, GA; [5]Dana-Farber Cancer Institute, Boston, MA; [6]Syndax Pharmaceuticals, Inc, Waltham, MA; [7]Merck & Co, Inc, Kenilworth, NJ; [8]Syndax Pharmaceuticals, Inc., New York, NY; [9]Memorial Sloan Kettering Cancer Center, New York, NY

BACKGROUND

- Single-agent programmed cell death-(ligand) 1 (PD-(L)1) therapies have profoundly improved treatment options for many cancers; still, a majority of patients do not respond[1]
- Resistance to immunotherapy may in part be due to an immunosuppressive tumor microenvironment (TME) in which cancer cells avoid detection and eradication by the host's immune system, mediated by[2,3]:
 - Upregulation of regulatory T cells, myeloid-derived suppressor cells (MDSCs), and indoleamine 2,3-dioxygenase in the TME
 - Low levels of tumor-infiltrating lymphocytes (CD8+ T cells) and tumor neoantigens
 - Low expression of immune-signaling molecules, such as PD-L1
 - T-cell apoptosis due to PD-(L)1 therapy-mediated increase in reactive oxygen species generation
- Entinostat (ENT) is an oral, class I selective histone deacetylase inhibitor[1]
 - ENT has demonstrated potent immunomodulatory activity by inhibition of MDSC function (**Figure 1**)
- ENCORE-601 phase 1b/2 study evaluates the safety and efficacy of ENT plus pembrolizumab (PEMBRO) in non-small cell lung cancer (NSCLC), melanoma, and mismatch repair-proficient colorectal cancer patients
 - Here, we report results in patients with NSCLC

Figure 1. **Immune Checkpoint Inhibitors and Entinostat Target Complementary Immunosuppression Mechanisms in the Tumor Microenvironment**

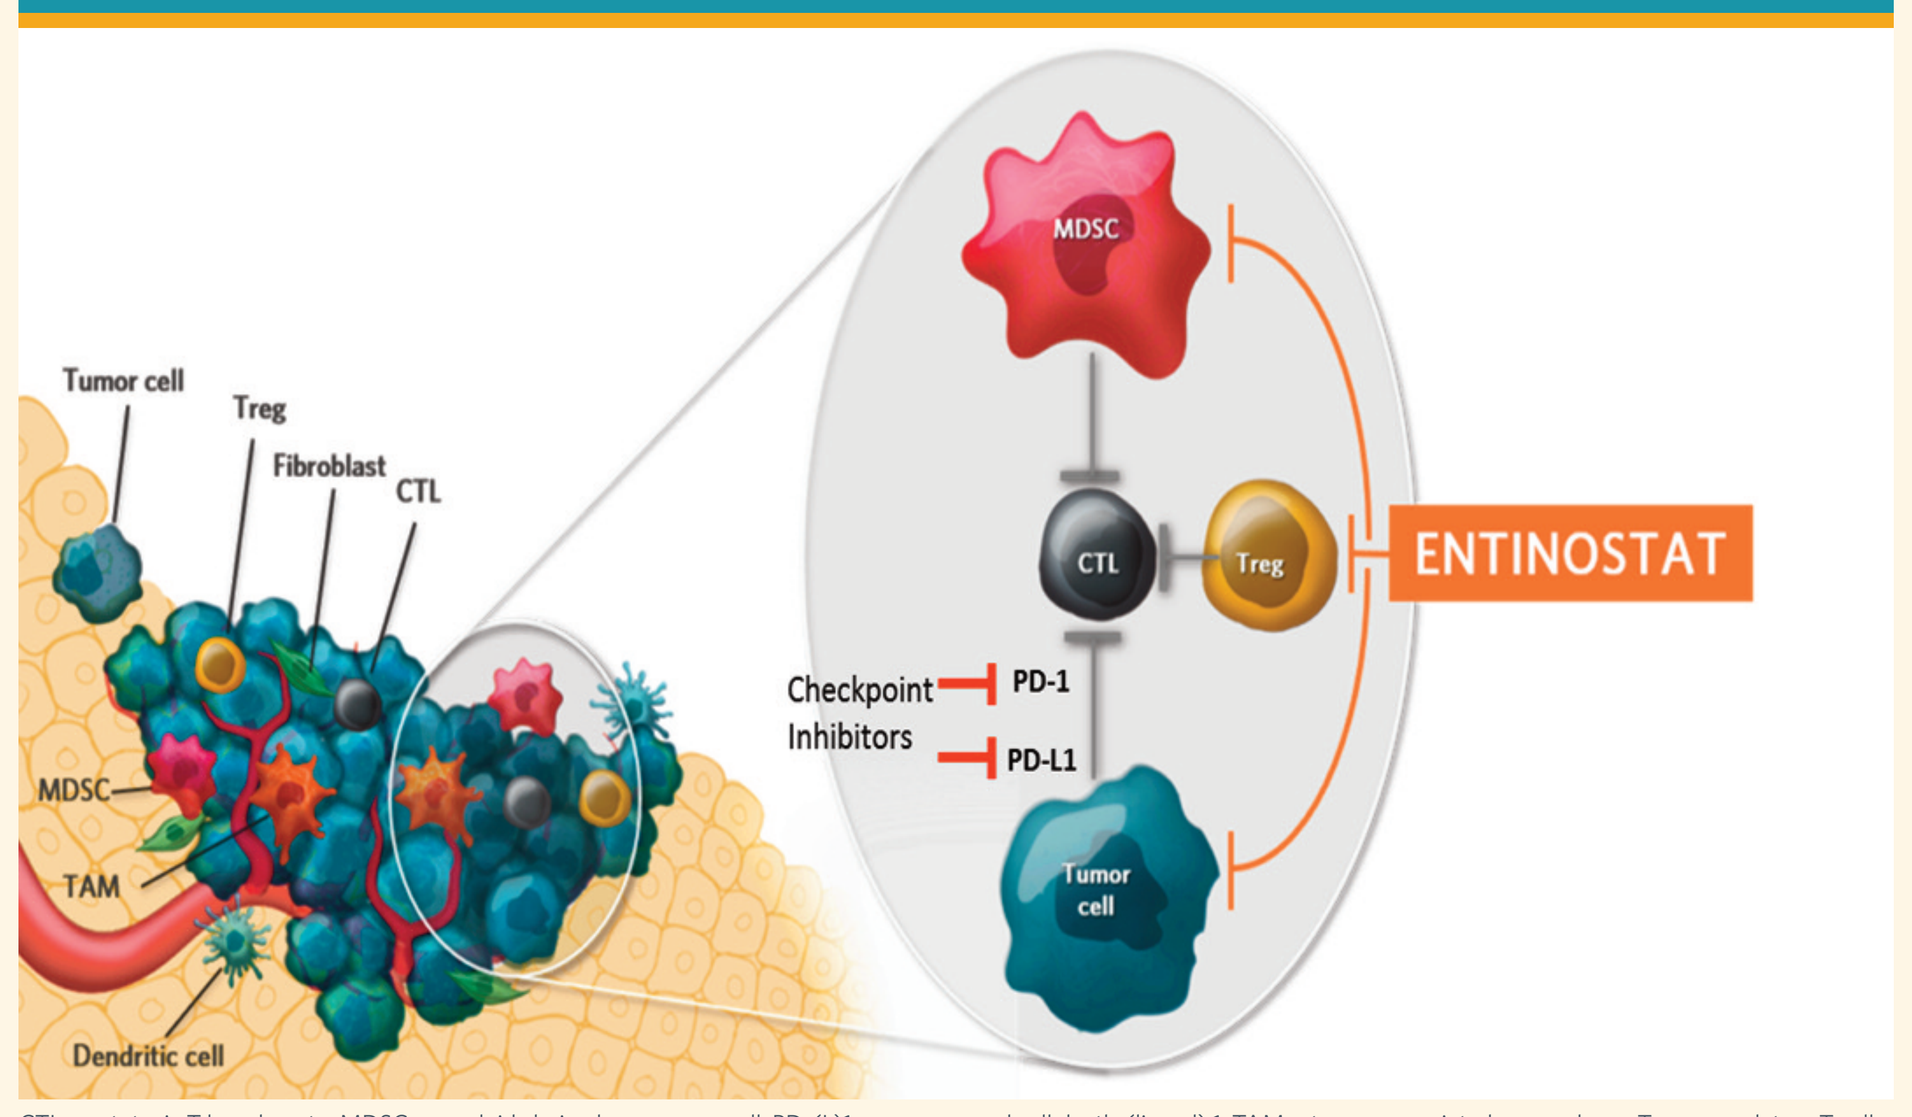
Tumor cell
Treg
Fibroblast
CTL
MDSC
TAM
Dendritic cell
MDSC
CTL
Treg
ENTINOSTAT
Checkpoint Inhibitors
PD-1
PD-L1
Tumor cell

CTL = cytotoxic T lymphocyte; MDSC = myeloid-derived suppressor cell; PD-(L)1 = programmed cell death-(ligand) 1; TAM = tumor-associated macrophage; Treg = regulatory T cell.

METHODS

Patients and Study Design

- Study design and objectives are summarized in **Figure 2**.

Figure 2. **Study Schema of Phase 1b/2 Simon 2-Stage Design**[4]

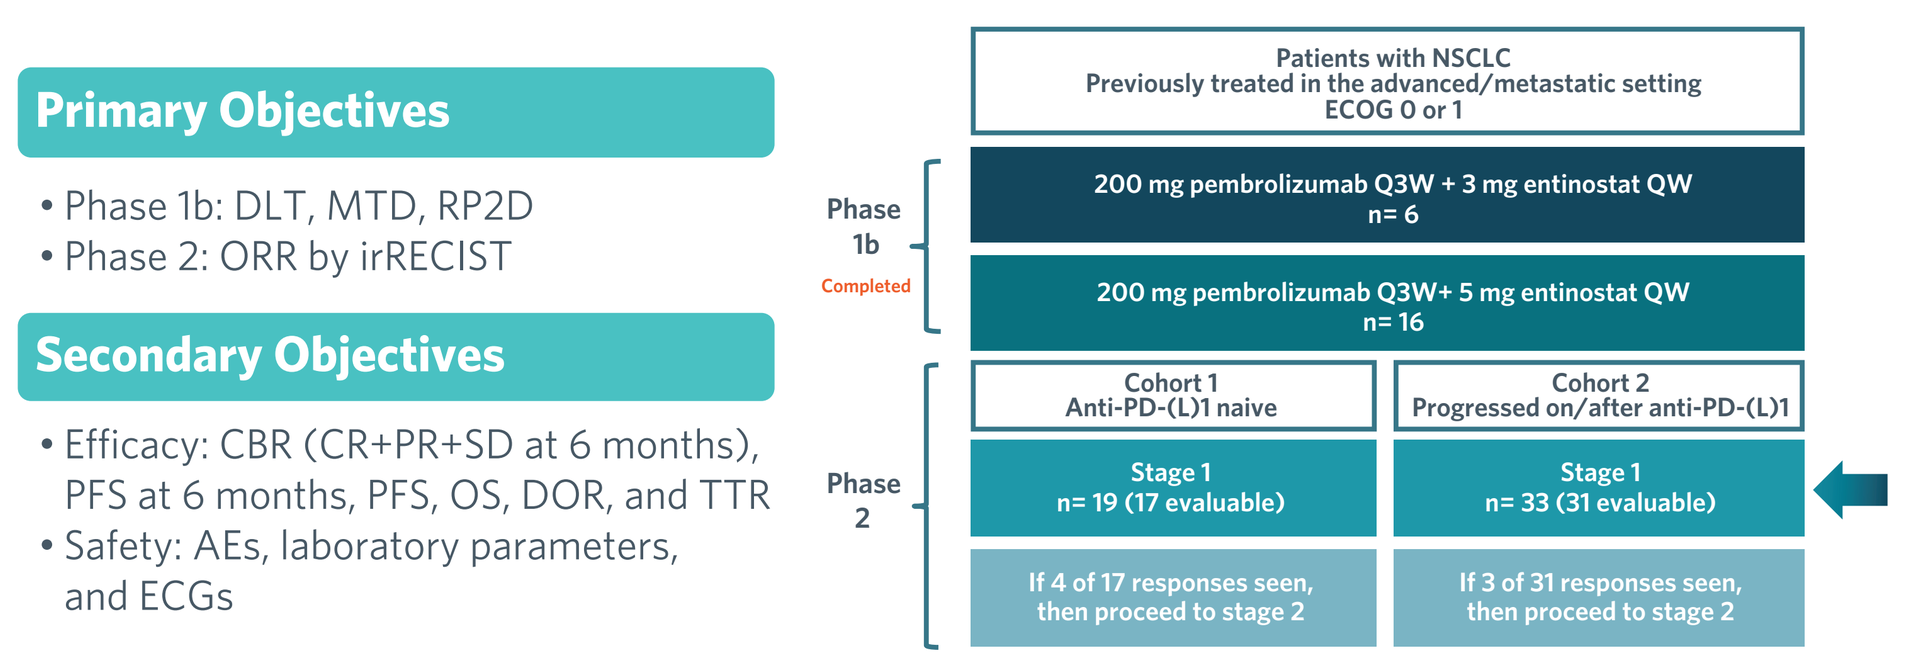
Primary Objectives
• Phase 1b: DLT, MTD, RP2D
• Phase 2: ORR by irRECIST
Secondary Objectives
• Efficacy: CBR (CR+PR+SD at 6 months), PFS at 6 months, PFS, OS, DOR, and TTR
• Safety: AEs, laboratory parameters, and ECGs
Patients with NSCLC Previously treated in the advanced/metastatic setting ECOG 0 or 1
Phase 1b
Completed
200 mg pembrolizumab Q3W + 3 mg entinostat QW n= 6
200 mg pembrolizumab Q3W+ 5 mg entinostat QW n= 16
Phase 2
Cohort 1 Anti-PD-(L)1 naive
Cohort 2 Progressed on/after anti-PD-(L)1
Stage 1 n= 19 (17 evaluable)
Stage 1 n= 33 (31 evaluable)
If 4 of 17 responses seen, then proceed to stage 2
If 3 of 31 responses seen, then proceed to stage 2

Evaluable patients defined as patients who reached the first tumor assessment timepoint or were discontinued for progression or adverse event prior to the first tumor assessment. AEs = adverse events; CBR = clinical benefit rate; CR = complete response; DLT = dose-limiting toxicities; DOR = duration of response; ECGs = electrocardiograms; ECOG = Eastern Cooperative Oncology Group; irRECIST = immune-related Response Evaluation Criteria in Solid Tumors; MTD = maximum tolerated dose; ORR = overall response rate; OS = overall survival; PFS = progression-free survival; PR = partial response; RP2D = recommended phase 2 dose; SD = stable disease; TTR = time to response.

Treatment and Assessments

- Patients received ENT 5 mg QW PO + PEMBRO 200 mg Q3W IV in 21-day cycles until disease progression or discontinuation for other reasons
- Response was assessed by RECIST v1.1 and irRECIST every 6 weeks
- Adverse events (AEs) were graded by National Cancer Institute Common Terminology Criteria for Adverse Events v4.03
- Peripheral blood and tumor tissue were collected for correlative studies (tumor PD-(L)1 expression and phenotypic and functional evaluation of immune cell subsets)

Statistical Assumptions

- The number of patients evaluated in each stage and the minimum number of responders needed to continue to the next stage were determined based on the optimum version of Simon's 2-stage design,[4] with 80% power and 1-sided significance level of 10%
 - Cohort 1: A true ORR of 35% for the combination is hypothesized, and the study is designed to exclude a lower bound of 20%
 - Cohort 2: A true ORR of 15% for the combination is hypothesized, and the study is designed to exclude a lower bound of 5%
- Inclusion of phase 1 patients:
 - Following the review of phase 1 data, the investigators and study sponsors jointly decided to include patients enrolled in the phase 1 portion who received 5 mg ENT into the phase 2 stage 1 assessment. This resulted in 7 patients being included in Cohort 1 and 9 patients included in Cohort 2. The stage 1 go/no-go criteria were adjusted accordingly

RESULTS

- Baseline demographic and prior PD-1 treatment history data are summarized in **Table 1** and **Table 2**

Table 1. **Baseline Demographics**

	Cohort 1 (n=19)	Cohort 2 (n=33)
Gender, n (%)		
Male/Female	11 (57.9)/8 (42.1)	17 (51.5)/16 (48.5)
Age (years)		
Median (range)	67.0 (43–78)	67.0 (48–86)
ECOG Performance Score, n (%)		
Grade 0/Grade 1	6 (31.6)/12 (63.2)	9 (27.3)/23 (69.7)
PD-L1 Expression, n (%)		
≥50% = Strong Positive	3 (15.8)	4 (12.1)
1%–49% = Weak Positive	5 (26.3)	13 (39.4)
<1% = Negative	6 (31.6)	15 (45.5)
Not Available	5 (26.3)	1 (3.0)
Smoking Status, n (%)		
Current	1 (5.3)	2 (6.0)
Former	13 (68.4)	29 (88.0)
Never	4 (21.0)	2 (6.0)
Missing	1 (5.3)	0

ECOG = Eastern Cooperative Oncology Group; PD-(L)1 = programmed cell death-(ligand) 1.

Table 2. **Prior PD-(L)1 History**

PD-(L)1 history	Cohort 2 (n=33)
Best Response on Prior PD-(L)1 therapy, n (%)	
Partial Response	1 (3.0)
Stable Disease	18 (54.5)
Disease Progression	13 (39.4)
Unknown	1 (3.0)
Duration on Prior PD-(L)1 therapy (days)	
Median	211.0
Duration between last dose of prior PD-(L)1 therapy and first dose of ENCORE-601 study therapy (days)	
Median	75.0

PD-(L)1 = programmed cell death-(ligand) 1.

Efficacy in Cohort 1: Anti-PD-(L)1–Naive Group

- 4 PRs out of 17 evaluable patients (24% ORR, 95% CI: 7-50) as shown in **Figure 3** and **Figure 4**
 - 3 confirmed, 1 unconfirmed PR due to new pericardial effusion with malignant cells
 - 3 with low or moderate baseline PD-L1 expression, 1 with unknown PD-(L)1 expression

Figure 3. **Swimmer Plot of Time to Response and Time on Treatment in Cohort 1 (anti-PD-(L)1-naive group)**

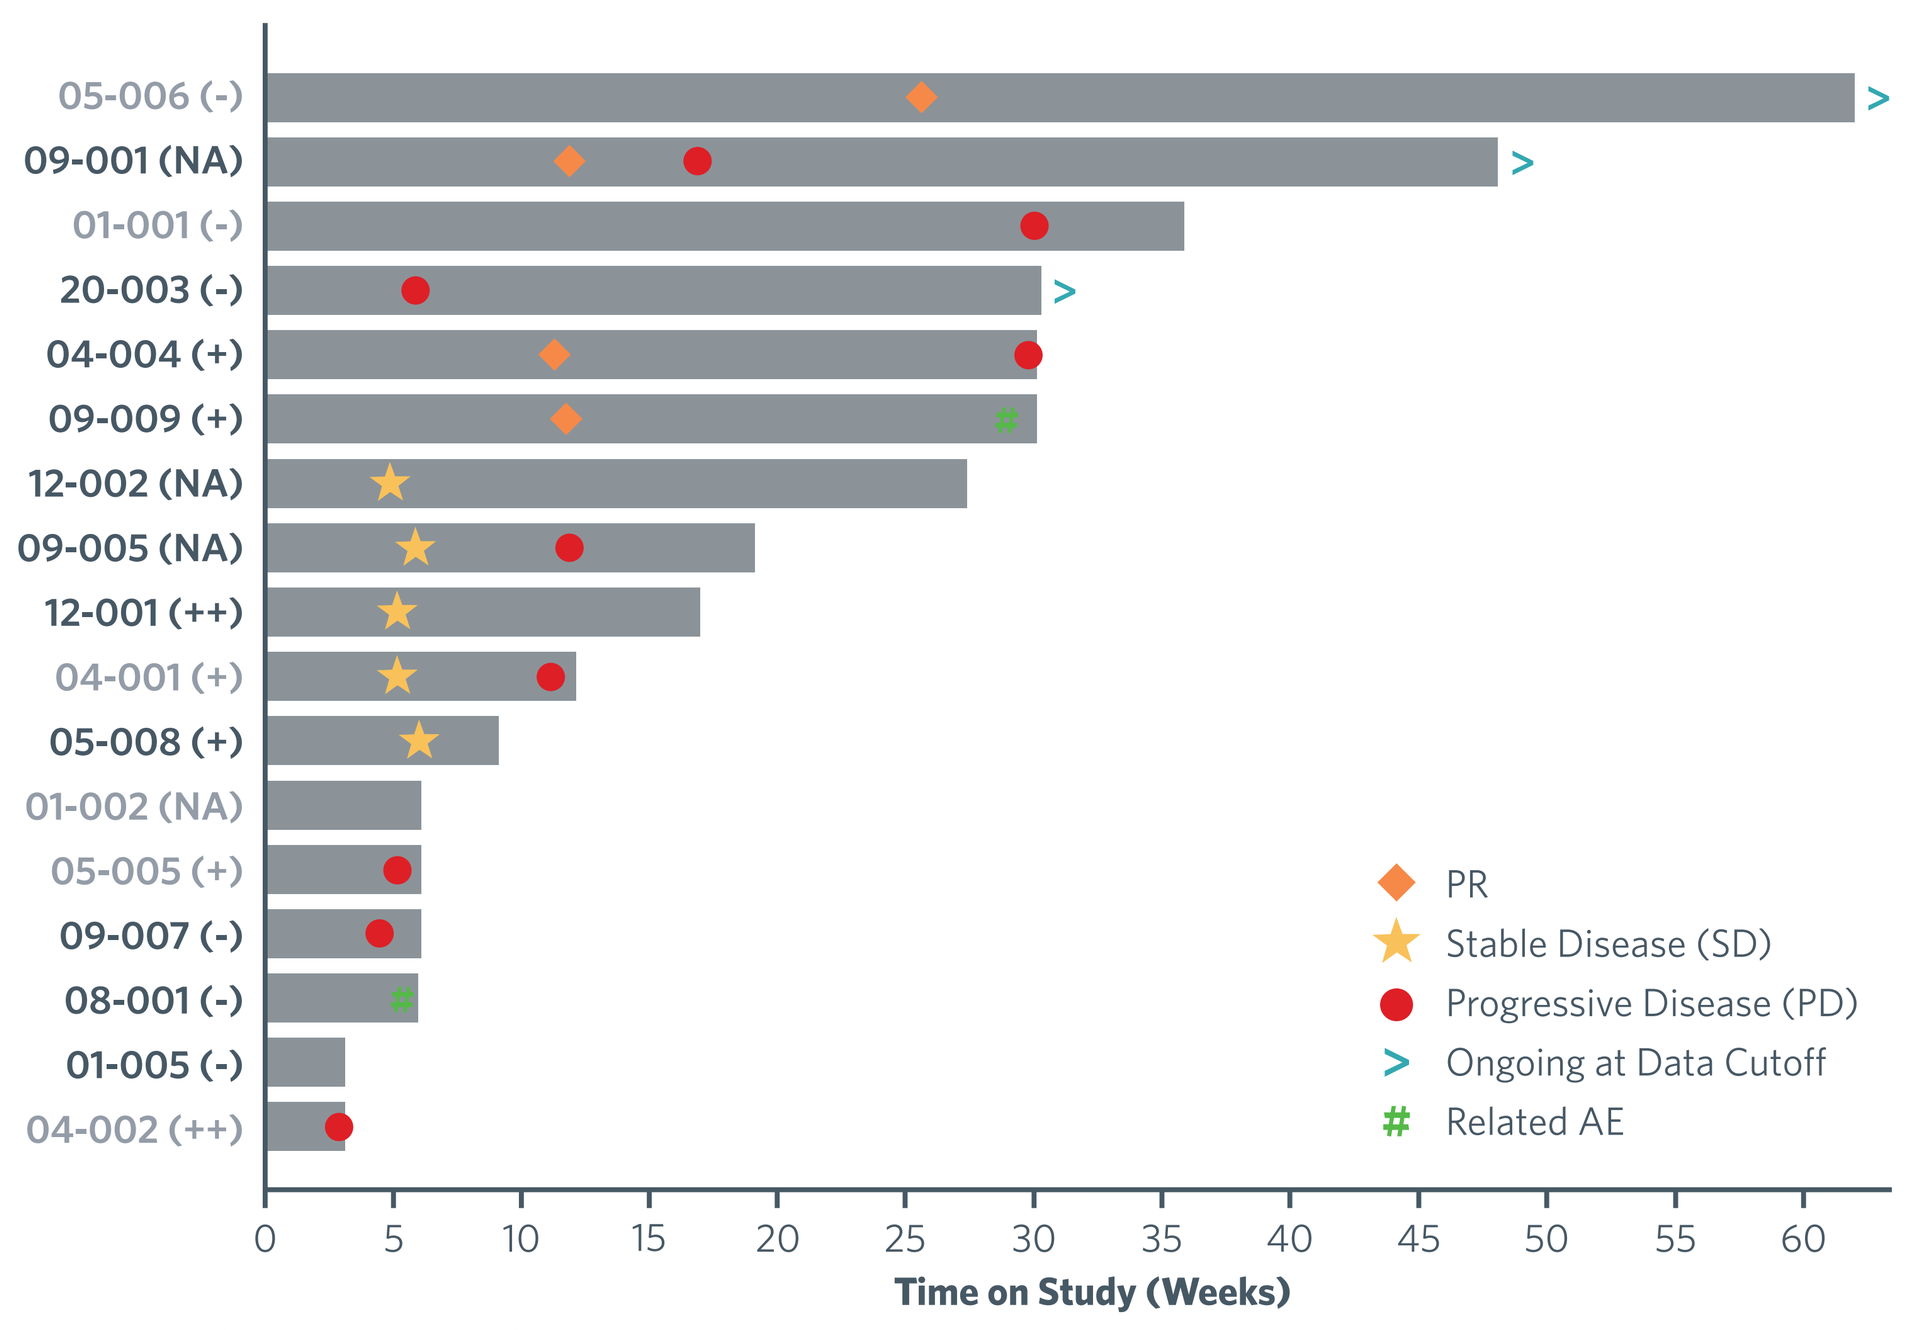
05-006 (-)
09-001 (NA)
01-001 (-)
20-003 (-)
04-004 (+)
09-009 (+)
12-002 (NA)
09-005 (NA)
12-001 (++)
04-001 (+)
05-008 (+)
01-002 (NA)
05-005 (+)
09-007 (-)
08-001 (-)
01-005 (-)
04-002 (++)
PR
Stable Disease (SD)
Progressive Disease (PD)
Ongoing at Data Cutoff
Related AE
0 5 10 15 20 25 30 35 40 45 50 55 60
Time on Study (Weeks)

PD-L1 expression: (-) = <1%, (+) = 1%-49%, (++) = ≥50%. Lighter gray = Phase 1 patients included in phase 2.

Figure 4. **Change in Tumor Size From Baseline in Cohort 1 (anti-PD-(L)1-naive group)**

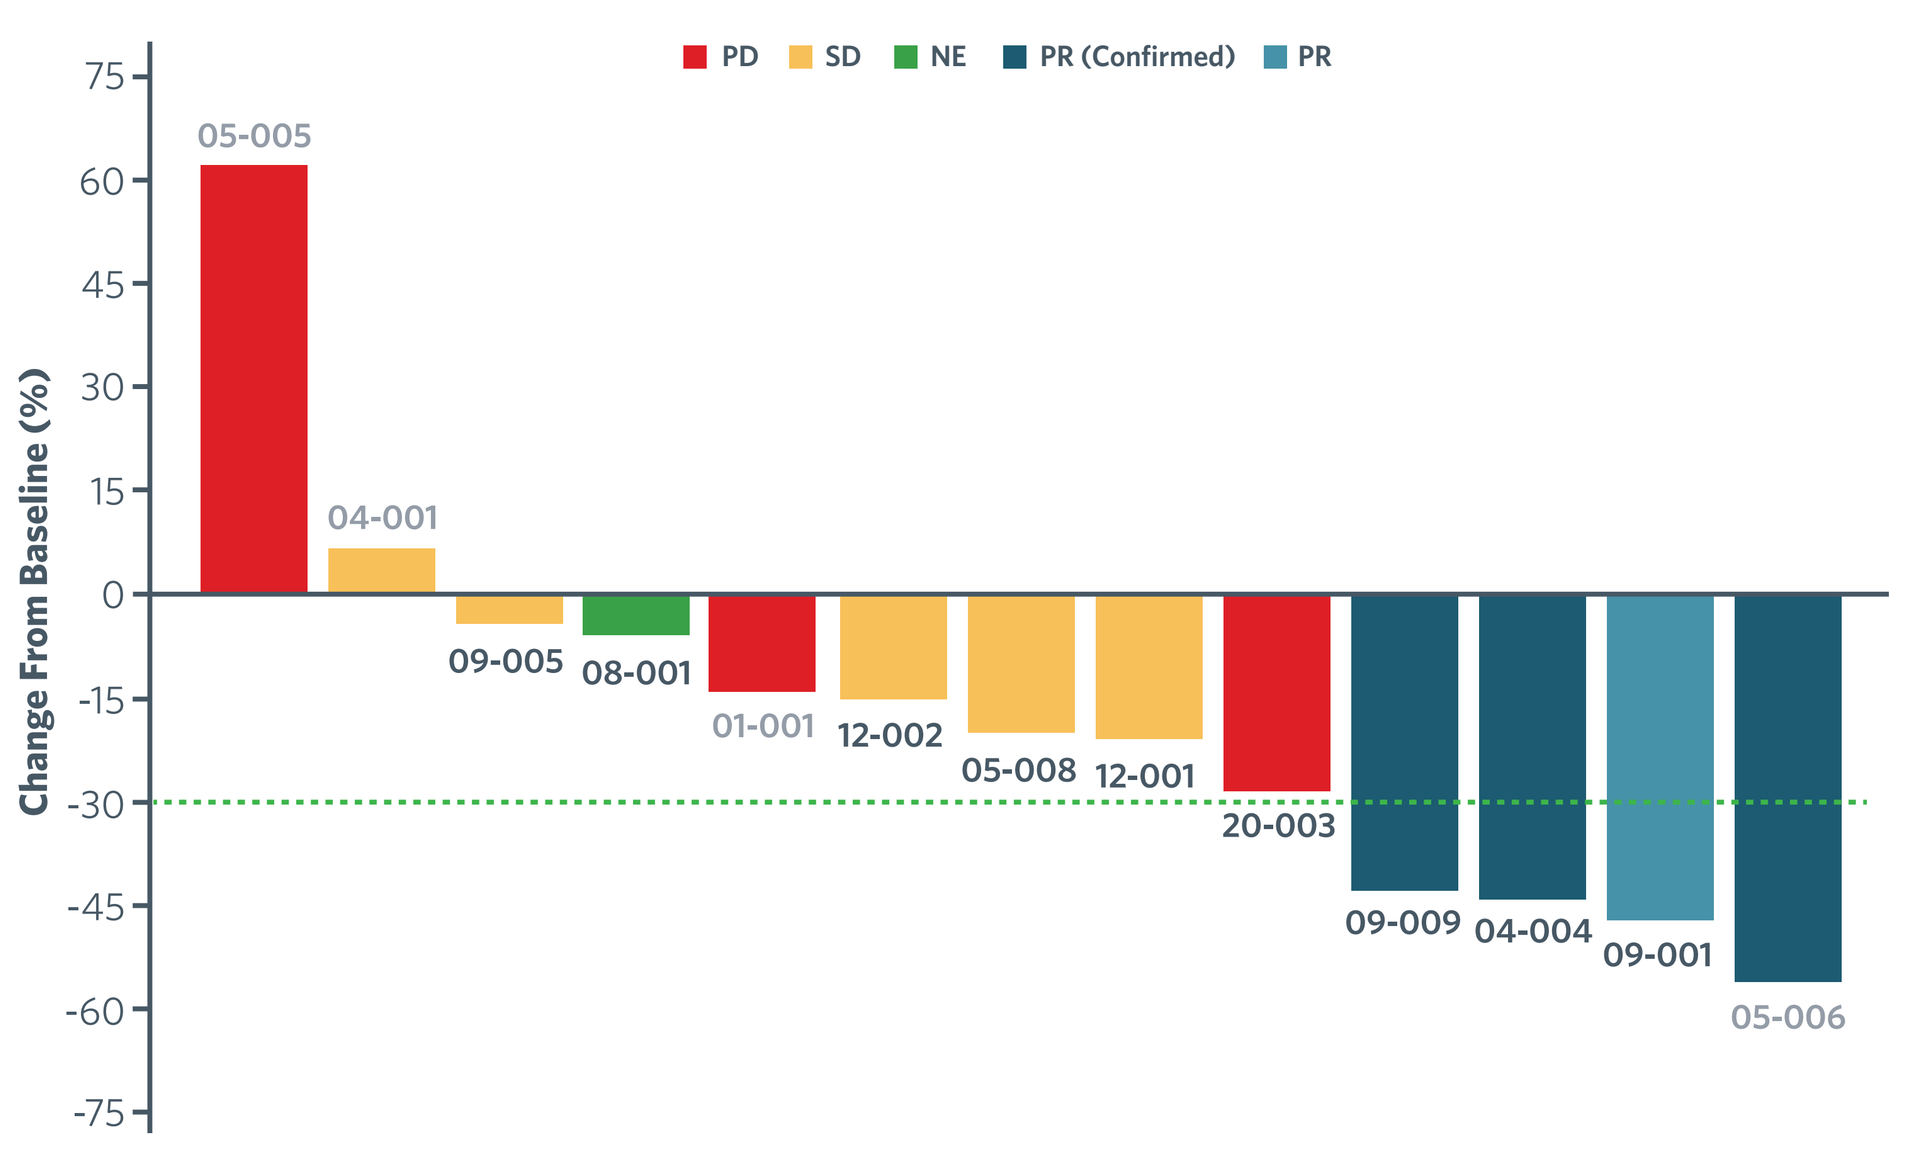
PD SD NE PR (Confirmed) PR
Change From Baseline (%)
05-005
04-001
09-005
08-001
01-001
12-002
05-008
12-001
20-003
09-009
04-004
09-001
05-006

Patients not on waterfall plot did not have any post-baseline tumor measurements. Lighter gray = Phase 1 patients included in phase 2.

Efficacy in Cohort 2: Progressed on/After Anti-PD-(L)1

- 3 PRs out of 31 evaluable patients (10% ORR, 95% CI: 2-26) as shown in **Figure 5**, **Figure 6**, and **Figure 7**
- 3 patients ongoing (1 PR, 2 SD)

RESULTS *Continued*

Figure 5. **Swimmer Plot of Time to Response and Time on Treatment in Cohort 2 (progressed on/after anti-PD-(L)1)**

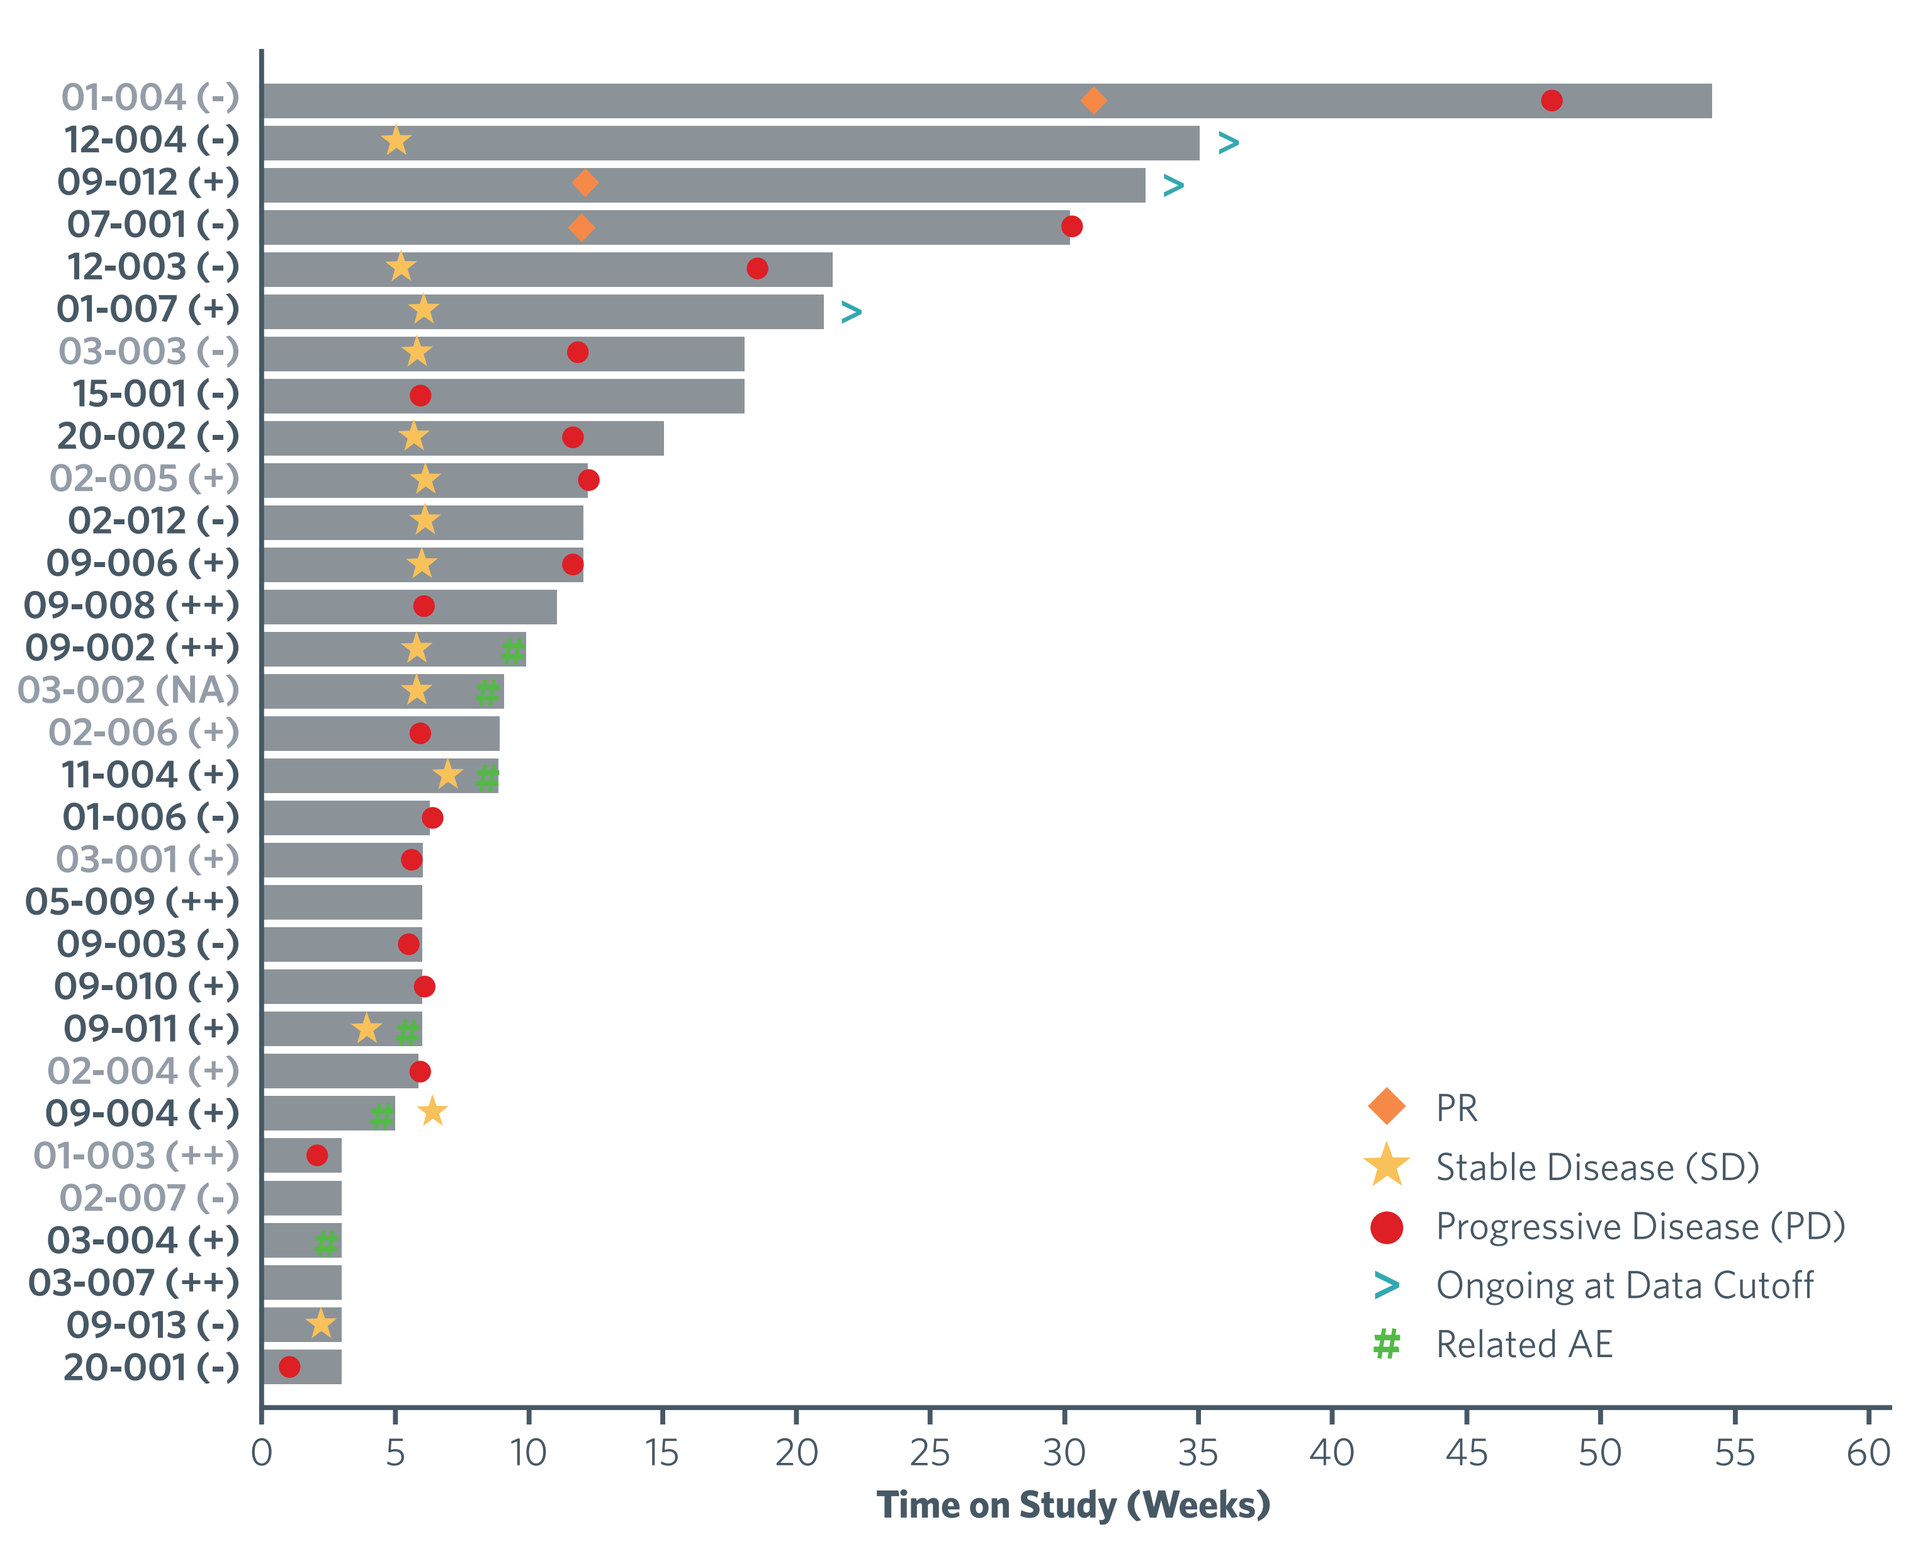
01-004 (-)
12-004 (-)
09-012 (+)
07-001 (-)
12-003 (-)
01-007 (+)
03-003 (-)
15-001 (-)
20-002 (-)
02-005 (+)
02-012 (-)
09-006 (+)
09-008 (++)
09-002 (++)
03-002 (NA)
02-006 (+)
11-004 (+)
01-006 (-)
03-001 (+)
05-009 (++)
09-003 (-)
09-010 (+)
09-011 (+)
02-004 (+)
09-004 (+)
01-003 (++)
02-007 (-)
03-004 (+)
03-007 (++)
09-013 (-)
20-001 (-)
PR
Stable Disease (SD)
Progressive Disease (PD)
Ongoing at Data Cutoff
Related AE
0 5 10 15 20 25 30 35 40 45 50 55 60
Time on Study (Weeks)

PD-L1 expression: (-) = <1%, (+) = 1%-49%, (++) = ≥50%. Lighter gray = Phase 1 patients included in phase 2.

Figure 6. **Change in Tumor Size From Baseline in Cohort 2 (progressed on/after anti-PD-(L)1)**

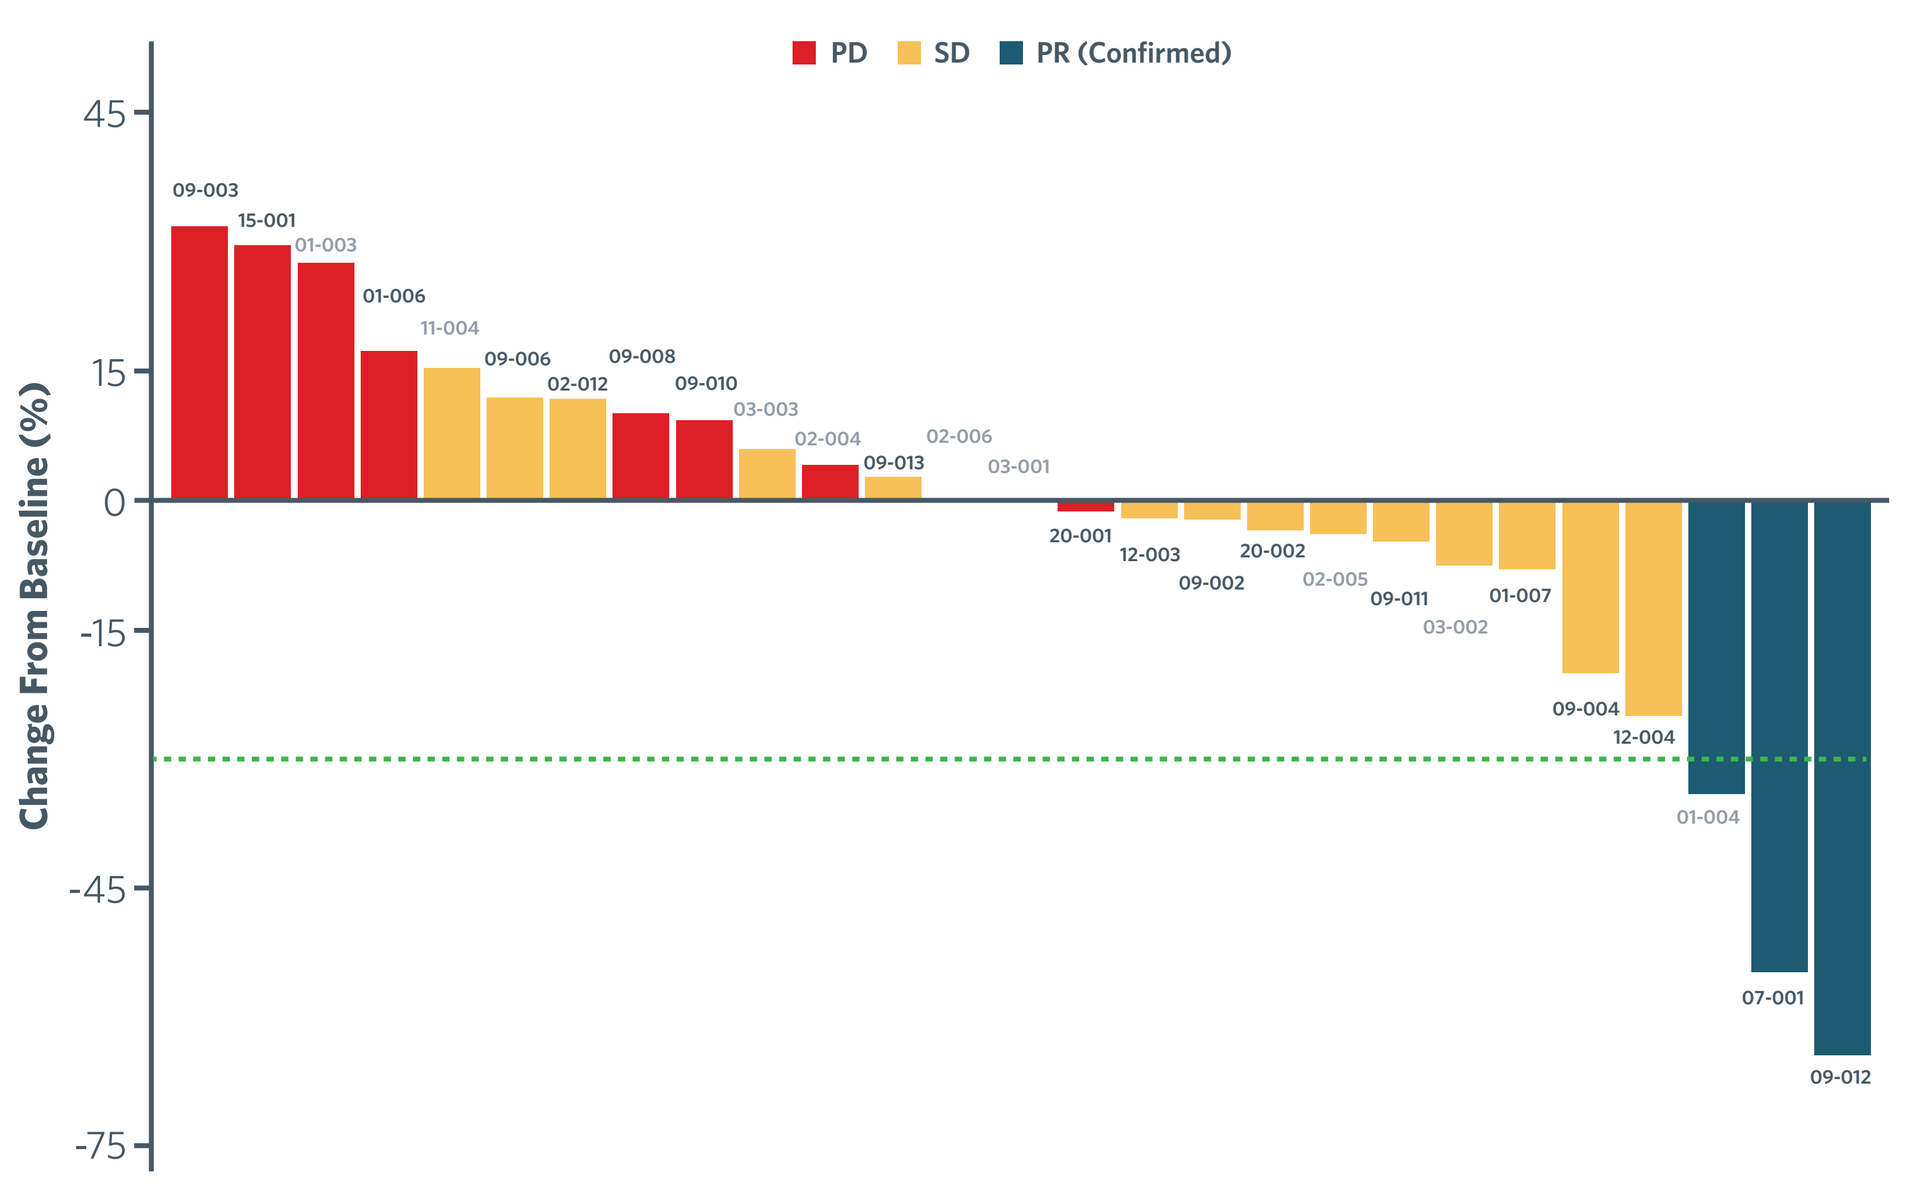
PD SD PR (Confirmed)
Change From Baseline (%)
09-003
15-001
01-003
01-006
11-004
09-006
02-012
09-008
09-010
03-003
02-004
09-013
02-006
03-001
20-001
12-003
09-002
20-002
02-005
09-011
03-002
01-007
09-004
12-004
01-004
07-001
09-012

Patients not on waterfall plot did not have any post-baseline tumor measurements. Lighter gray = Phase 1 patients included in phase 2.

Figure 7. **Spider Plot of Percentage Change in Tumor Size Relative to Baseline by Weeks From Start of Study in Responders in Cohort 2 (sum of diameters of target lesions)**

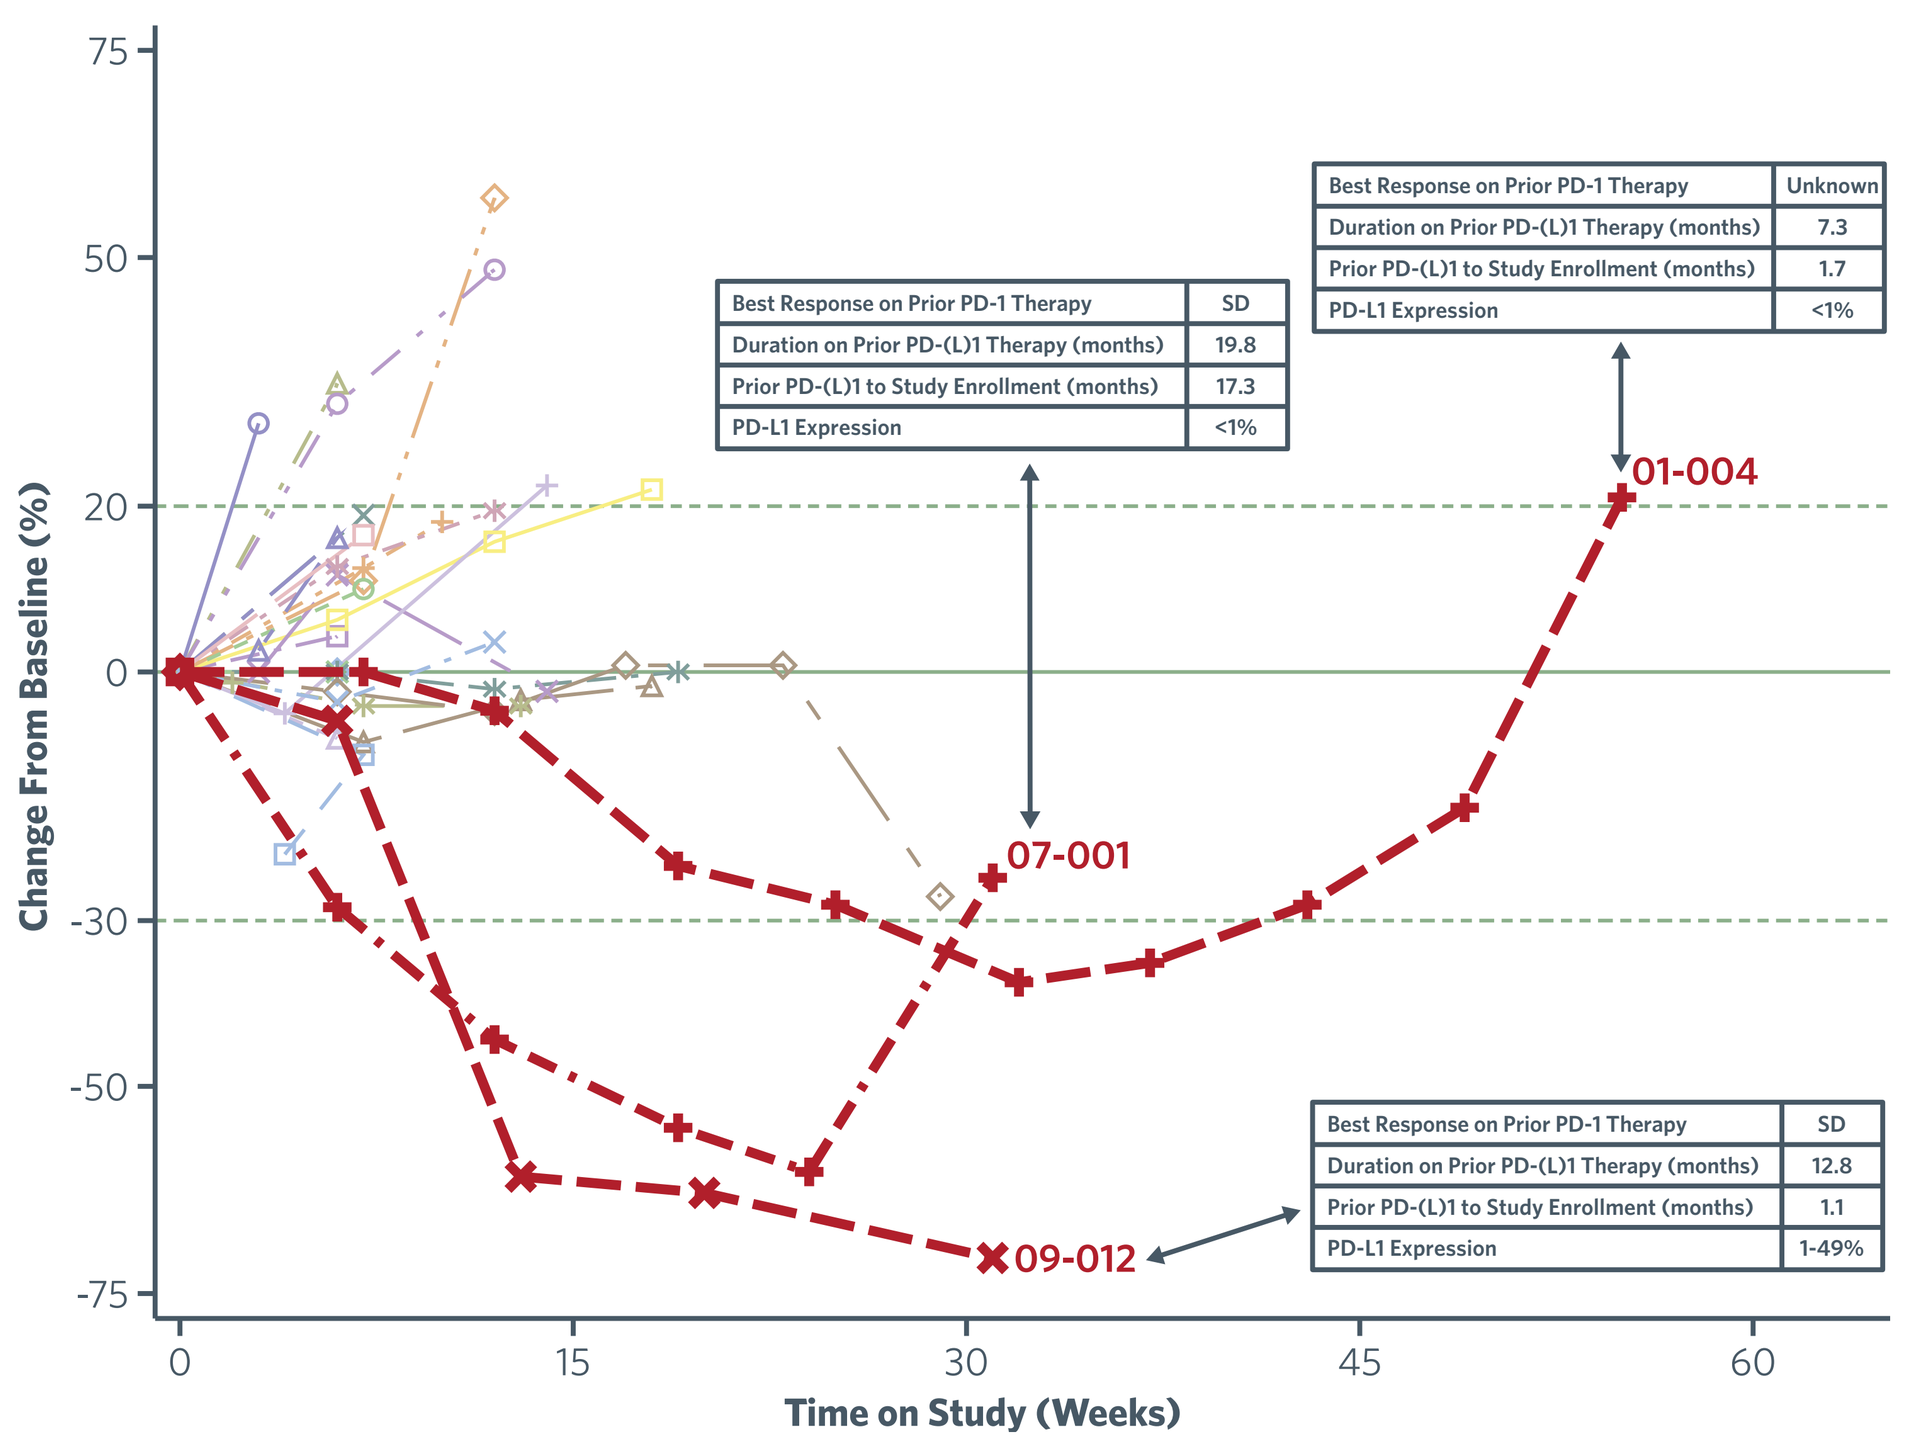
Best Response on Prior PD-1 Therapy | Unknown
Duration on Prior PD-(L)1 Therapy (months) | 7.3
Prior PD-(L)1 to Study Enrollment (months) | 1.7
PD-L1 Expression | <1%
Best Response on Prior PD-1 Therapy | SD
Duration on Prior PD-(L)1 Therapy (months) | 19.8
Prior PD-(L)1 to Study Enrollment (months) | 17.3
PD-L1 Expression | <1%
Best Response on Prior PD-1 Therapy | SD
Duration on Prior PD-(L)1 Therapy (months) | 12.8
Prior PD-(L)1 to Study Enrollment (months) | 1.1
PD-L1 Expression | 1-49%
01-004
07-001
09-012
Change From Baseline (%)
Time on Study (Weeks)

RESULTS *Continued*

Safety

- 40.4% of patients experienced at least one Grade ≥3 event deemed related to study drug as shown in **Table 3**

Table 3. **Safety: Treatment-Related Adverse Events (>10%)**

	Cohort 1 (n=19)		Cohort 2 (n=33)		Total (N=52)	
	All Grades	Grade ≥3	All Grades	Grade ≥3	All Grades	Grade ≥3
Subjects With At Least One Related Adverse Event, n (%)	15 (78.9)	6 (31.6)	27 (81.8)	15 (45.5)	42 (80.8)	21 (40.4)
Fatigue	12 (63.2)	0	14 (42.4)	4 (12.1)	26 (50.0)	4 (7.7)
Anemia	4 (21.1)	1 (5.3)	8 (24.2)	4 (12.1)	12 (23.1)	5 (9.6)
Decreased appetite	2 (10.5)	0	7 (21.2)	0	9 (17.3)	0
Diarrhea	3 (15.8)	0	6 (18.2)	1 (3.0)	9 (17.3)	1 (1.9)
Platelet count decreased	3 (15.8)	0	5 (15.2)	0	8 (15.4)	0
Pruritus	4 (21.1)	0	3 (9.1)	0	7 (13.5)	0
Nausea	3 (15.8)	0	3 (9.1)	0	6 (11.5)	0
Pneumonitis	2 (10.5)	1 (5.3)	4 (12.1)	3 (9.1)	6 (11.5)	4 (7.7)

- 7 patients experienced Grade ≥3 immune-related adverse event as shown in **Table 4**
 - 1 in Cohort 1 (5%) and 6 in Cohort 2 (18%)
 - 1 patient in Cohort 2 experienced 2 irAEs
- 5 patients discontinued due to these AEs
 - 1 in Cohort 1 and 4 in Cohort 2

Table 4. **Safety: Grade ≥3 Immune-Related Adverse Events**

	Cohort 1 (n=19)	Cohort 2 (n=33)	Total (N = 52)
Pneumonitis, n (%)	1 (5.3)	3 (9.1)	4 (7.7)
Colitis, n (%)	0	2 (6.1)	2 (3.8)
Encephalitis, n (%)	0	1 (3.0)	1 (1.9)
Hyperthyroidism, n (%)	0	1 (3.0)	1 (1.9)

Correlative Studies

- MDSCs were measured at cycle 1, day 1 (C1D1) and cycle 2, day 15 (C2D15) in Cohort 1 (n=8) and Cohort 2 (n=13) as shown in **Figure 8**
 - Cohort 1: all MDSC subsets decreased
 - early-stage (e)MDSCs (−56.5%); polymorphonuclear (PMN)-MDSCs (−60.3%); monocyte (M)-MDSCs (−72.3%)
 - Cohort 2:
 - eMDSCs (−53.3%); PMN-MDSCs (+1.0%); M-MDSCs (−45.6%)
 - eMDSCs and M-MDSCs decreased in all responders (5/5) and PMN-MDSCs in 4/5

Figure 8. **Combination Resulted in General Decreases in MDSCs**

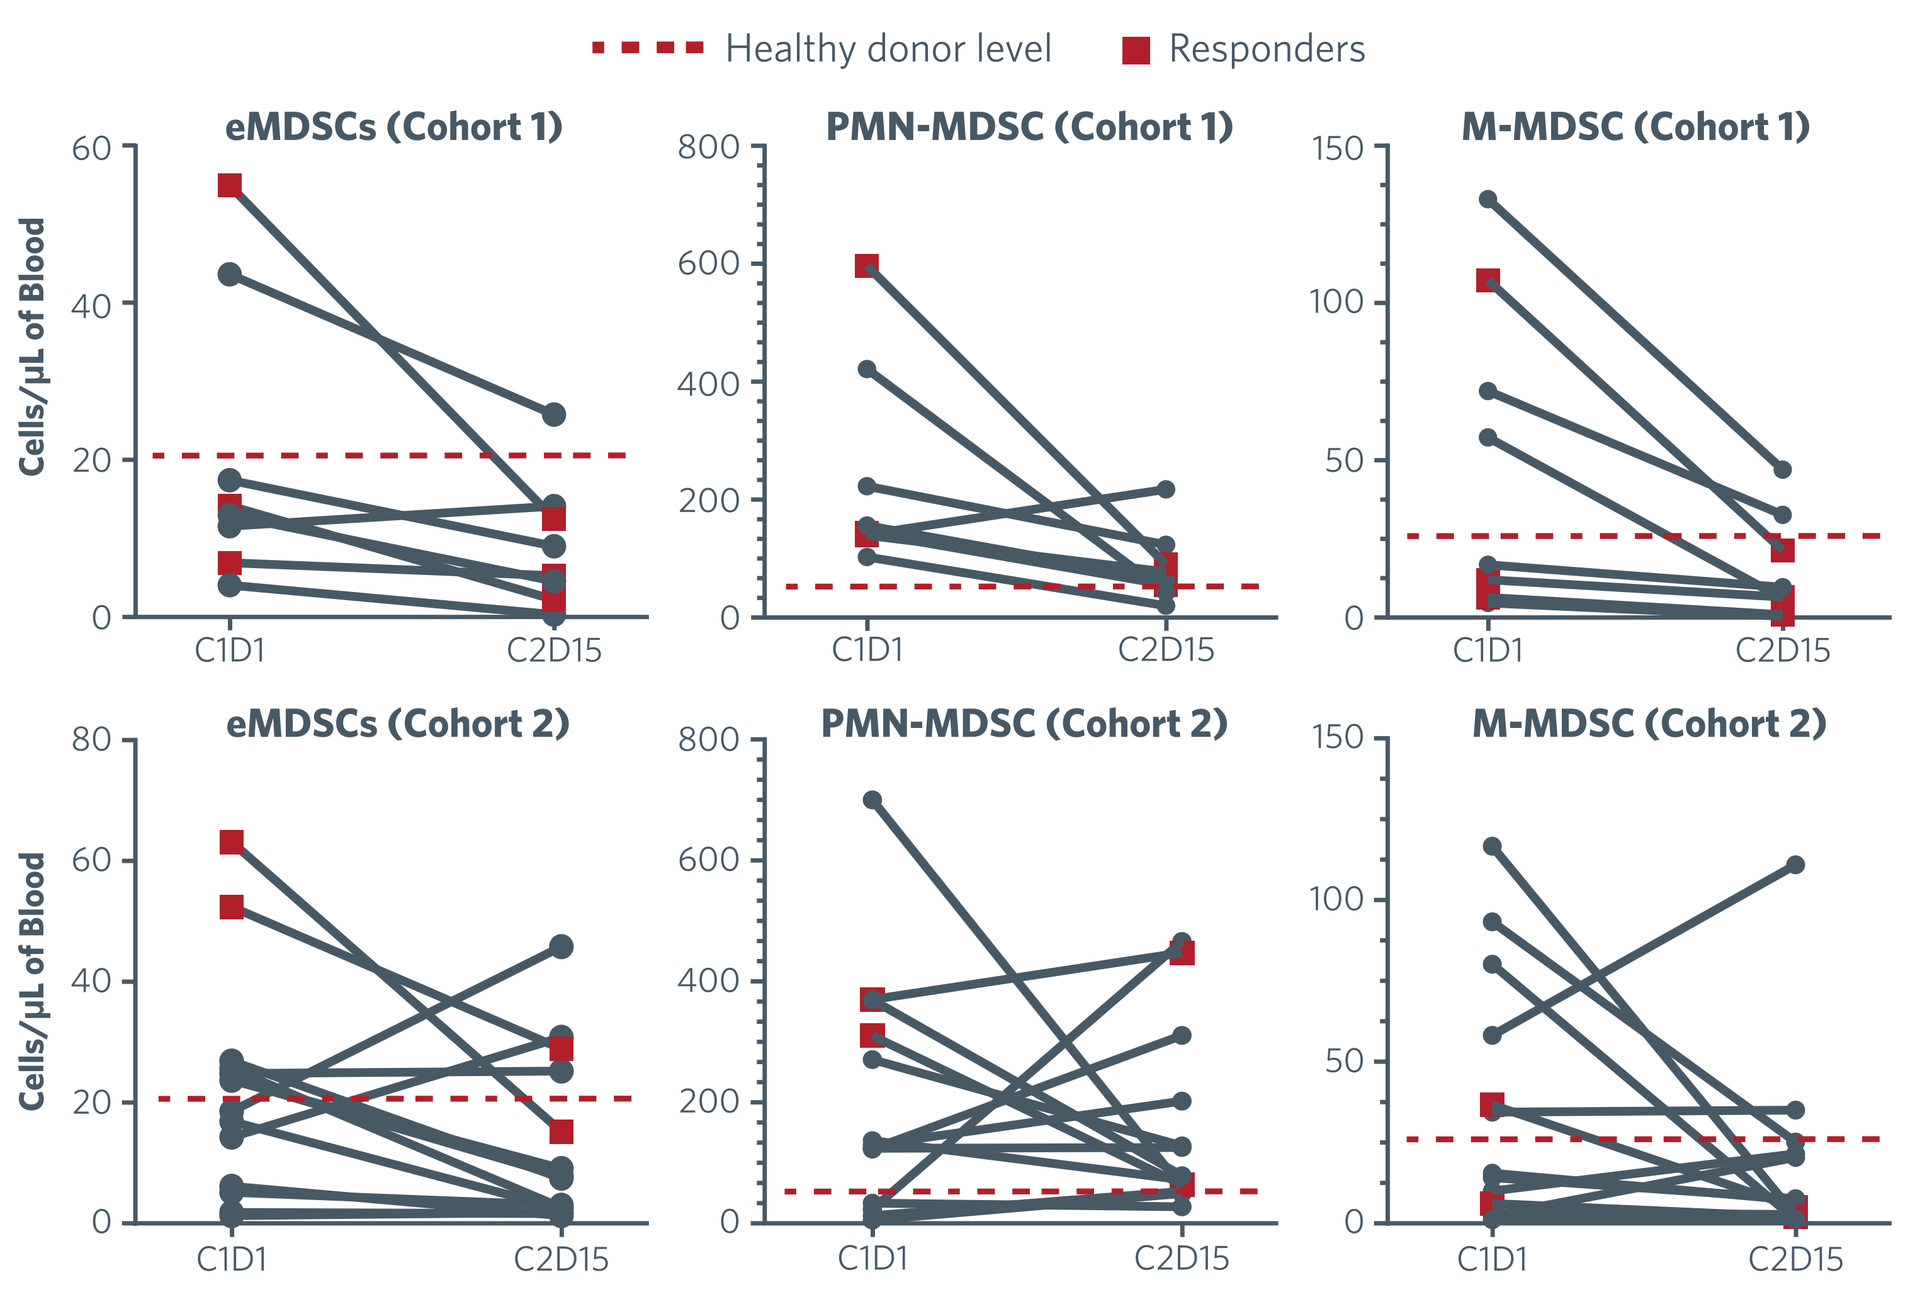
Healthy donor level
Responders
eMDSCs (Cohort 1)
PMN-MDSC (Cohort 1)
M-MDSC (Cohort 1)
eMDSCs (Cohort 2)
PMN-MDSC (Cohort 2)
M-MDSC (Cohort 2)
Cells/μL of Blood
C1D1
C2D15

eMDSC, early-stage MDSC; M-MDSC, monocytic-MDSC; MDSC, myeloid-derived suppressor cells; PBMC, peripheral blood mononuclear cell; PMN-MDSC; polymorphonuclear-MDSC.

CONCLUSIONS

- Combination ENT and PEMBRO demonstrates antitumor activity
 - 24% ORR in anti-PD-(L)1–naive patients
 - 10% ORR in patients who progressed on prior PD-(L)1 blockade
 - Responses seen in patients with negative to low PD-L1 expression
- Acceptable safety in patients with NSCLC who are both naive to and have progressed on prior PD-(L)1 blockade
 - Potential increase in immune-related toxicity in those who had progressed on prior PD-(L)1 therapy. Additional data from stage 2 will further elucidate
- Reductions in circulating myeloid-derived suppressor cells were observed following treatment
- Cohort 2 has advanced to stage 2 and is currently enrolling
 - Additional patients are not being enrolled in Cohort 1 at this time

References

1. Orillion A et al. *Clin Cancer Res.* 2017;23(17):5187-5201; **2.** Pitt JM et al. *Immunity.* 2016;44(6): 1255-1269; **3.** Tkachev V et al. *J Immunol.* 2015;194(12):5789-5800; **4.** Simon R. *Oncology (Williston Park).* 1989;3(7):43-49.

Acknowledgments

The authors thank the patients and their families, investigators and study staff, and The Wistar Institute. This study was sponsored by Syndax Pharmaceuticals, Inc., in collaboration with Merck & Co., Inc., Kenilworth, NJ. Writing and editorial assistance was provided by Adelphi Communications, New York. This assistance was funded by Syndax Pharmaceuticals, Inc.

The QR code is intended to provide scientific information for individual reference. The PDF should not be altered or reproduced in any way.

Presented at the Society for the Immunotherapy of Cancer (SITC) Annual Meeting; November 8–12, 2017; National Harbor, MD, USA